Neuropilin-2, The Specific Binding Partner To ATYR1923, is Expressed In Sarcoid Granulomas and Key Immune Cells

Suzanne Paz, Dalena Chu, Michaela Ferrer, Kendall Walwick, Clara Polizzi, Jeanette Ampudia, Steve Crampton, Christoph Burkart, Leslie Nangle, Sanna Rosengren

aTyr Pharma, San Diego, California, USA

Abstract

Neuropilin-2 (NRP2) is a pleiotropic co-receptor with an emerging role in the regulation of inflammatory responses. NRP2 was identified by aTyr as the sole binding partner for ATYR1923, a fusion protein combining a novel immunomodulatory domain from histidyl-tRNA synthetase (HARS) and a human IgG1 Fc. ATYR1923 is in early clinical testing for pulmonary sarcoidosis, an inflammatory condition that can lead to lung fibrosis. To date, little is known about the role of NRP2 in immune regulation and disease, in particular very little is known about the expression of NRP2 in sarcoidosis patients. We sought to characterize NRP2 expression patterns on immune cells implicated in the pathology of sarcoidosis.

Using RNAscope® technology, we demonstrate that NRP2 mRNA is present in both skin and lung samples obtained from sarcoidosis patients. NRP2 expression was readily detectable in granulomas with positive signals predominantly localized within the inner structure of the granuloma where Langham's giant cells (myeloid origin) reside. This finding was confirmed in both lung and skin samples from sarcoidosis patients. We thus sought to investigate which immune cells express NRP2.

In vitro, we show that NRP2 is highly inducible in key immune cells following immunomodulatory insults such as lipopolysaccharide (LPS) or other toll-like-receptor ligands (TLR). Interestingly, NRP2 expression on macrophages was only induced following activation of toll-like receptors found on the cell surface (TLR1, TLR2, TLR4, TLR5, TLR6), but not by endosomal TLR ligands (TLR3, TLR7/8, TLR9). In addition to detecting NRP2 on the cell surface of macrophages, we also confirm its presence on dendritic cells, T cells and T regulatory cells using either cell lines or primary cells.

In vivo, multi-color flow cytometry was used to immunophenotype either lungs or spleens isolated from LPS challenged animals. We confirmed our *in vitro* findings demonstrating that expression of NRP2 is present and inducible on the cell surface of various immune cells (macrophages, dendritic cells, neutrophils, and T cells).

We report for the first time that NRP2 is expressed in samples obtained from lung and skin of sarcoidosis patients. We clearly show that NRP2 expression can be detected on key immune cells known to play an important role in inflammation and granuloma formation. These findings highlight the potential of ATYR1923 to exert its effect on various immune cells directly related to the pathology of the target patient population.

Introduction

- Sarcoidosis is an inflammatory disorder of unknown etiology
- The disease involves granuloma formation (clumps of inflammatory cells) in affected organs, which can lead to fibrosis and irreversible organ damage. Lungs are the most commonly affected organ (>90% of cases) and this is referred to as pulmonary sarcoidosis
- We previously demonstrated that ATYR1923, a fusion protein combining a novel immunomodulatory domain from histidyl-tRNA synthetase (HARS) and a human IgG1 Fc binds to the NRP2 receptor in human and mouse overexpressing cells (Paz et al., Keystone 2019; Xu et al., ATS 2020)
- Little is known about NRP2's role in immune regulation and disease/granuloma progression

Figure 1. Granuloma Formation in Pulmonary Sarcoidosis

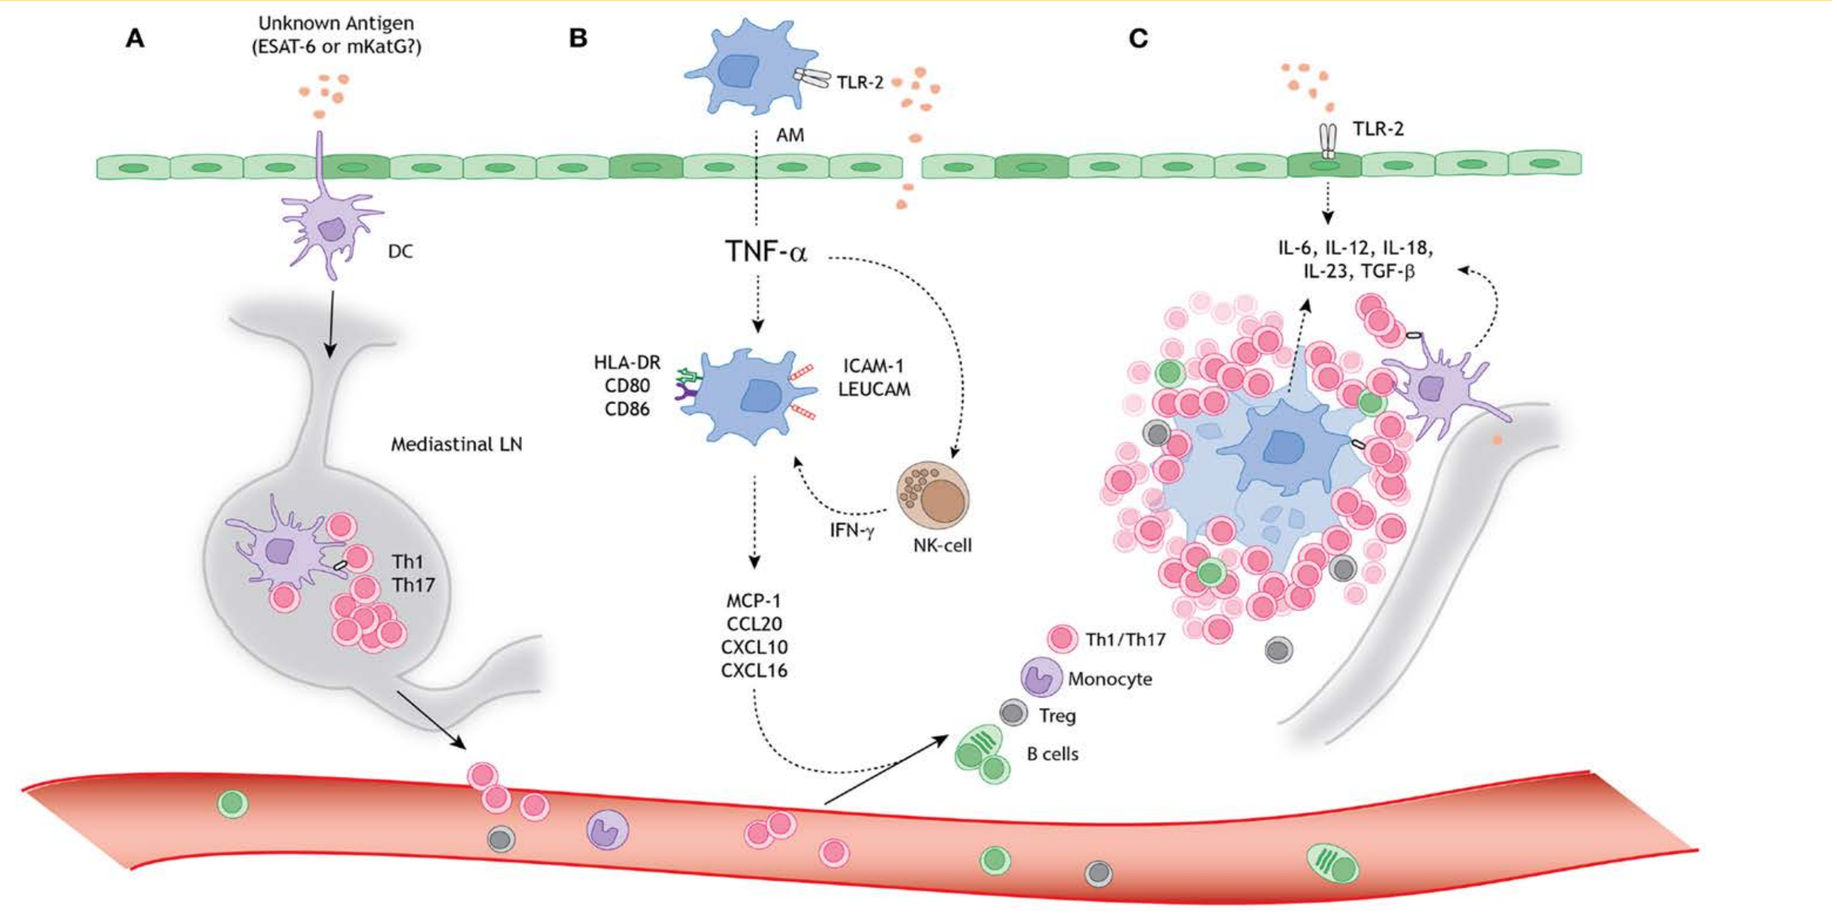
A
Unknown Antigen (ESAT-6 or mKatG?)
DC
Mediastinal LN
Th1
Th17
B
TLR-2
AM
TNF-α
HLA-DR
CD80
CD86
ICAM-1
LEUCAM
IFN-γ
NK-cell
MCP-1
CCL20
CXCL10
CXCL16
C
TLR-2
IL-6, IL-12, IL-18, IL-23, TGF-β
Th1/Th17
Monocyte
Treg
B cells

Figure 1. An unknown antigen activates **(A)** interstitial DCs, **(B)** alveolar macrophages (AMs), and **(C)** alveolar epithelial cells type II (AEC-II). This process is possibly initiated by TLR-2 ligands. **(A)** The interstitial DCs pick up the antigen and migrate toward the mediastinal lymph nodes (LNs), where they initiate differentiation and clonal expansion of T helper (Th)1 and 17 ligands under stimulation of both TNF-α and natural-killer (NK) cell-derived INF-γ, thereby attracting Th1/17 cells, monocytes, regulatory T cells (Tregs), and B cells. **(C)** The lung environment is characterized by the presence of Th1 and Th17 favoring cytokines produced by macrophages, DCs, and AEC-II. Persistent stimulation, mediated by APCs, leads to continuous cellular cells. **(B)** Simultaneously, AMs produce TNF-α, which initiates upregulation of activation (HLA-DR and CD80/86) and adhesion (ICAM-1 and LeuCAM) molecules. Macrophages produce chemokine recruitment to the site of inflammation, which leads to granuloma formation. Tregs infiltrating the granuloma fail to diminish the exaggerated immune response, thereby contributing to granuloma persistence and integrity.

Experimental Procedure

Figure 2. RNAscope® Methodology

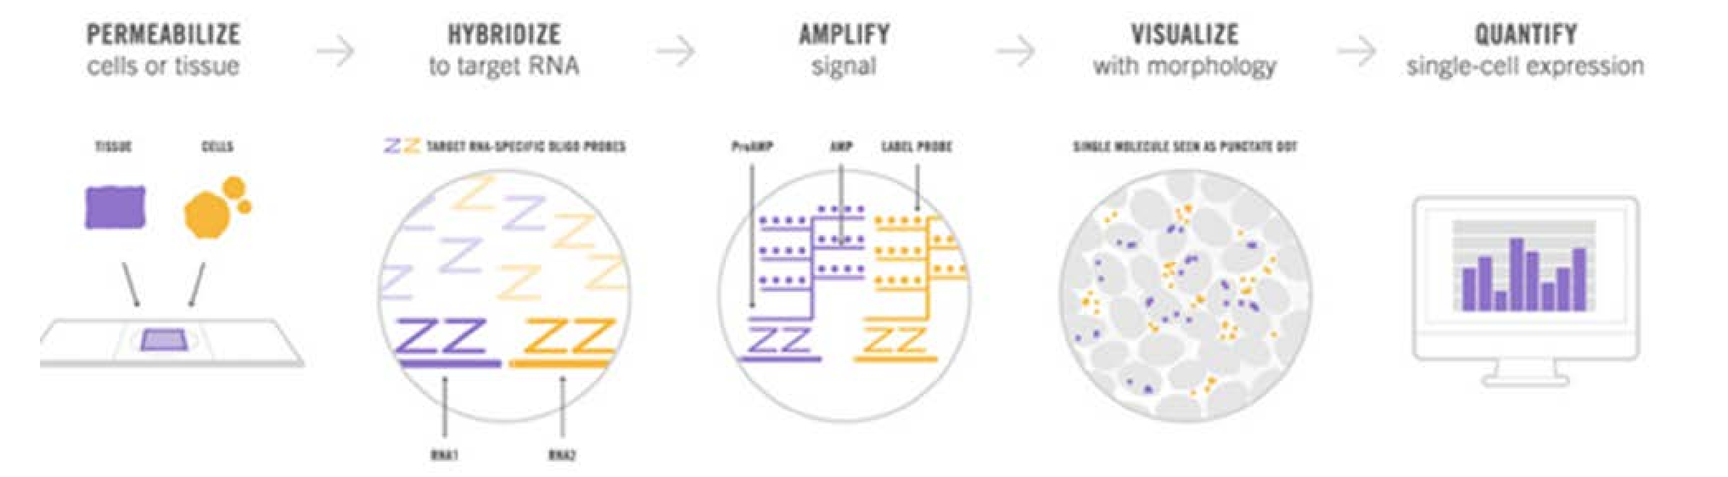
PERMEABILIZE
cells or tissue
HYBRIDIZE
to target RNA
AMPLIFY
signal
VISUALIZE
with morphology
QUANTIFY
single-cell expression

Figure 2. Schematic Representation showing the experimental procedures utilized to measure NRP2 mRNA expression in skin and lung biopsy samples. Briefly, fixed tissue sections on slides were pretreated with a pretreatment kit to block endogenous peroxidase activity and optimally permeabilize samples to allow probe access to target RNA. With approximately 20 specific double Z probe pairs per target, probes hybridize to target RNA molecules. Detection reagents amplify the hybridization signals via sequential hybridization of amplifiers and labeled probes. Each punctuated dot signal represents a positive signal.

Figure 3. Generation of Human Primary Macrophages

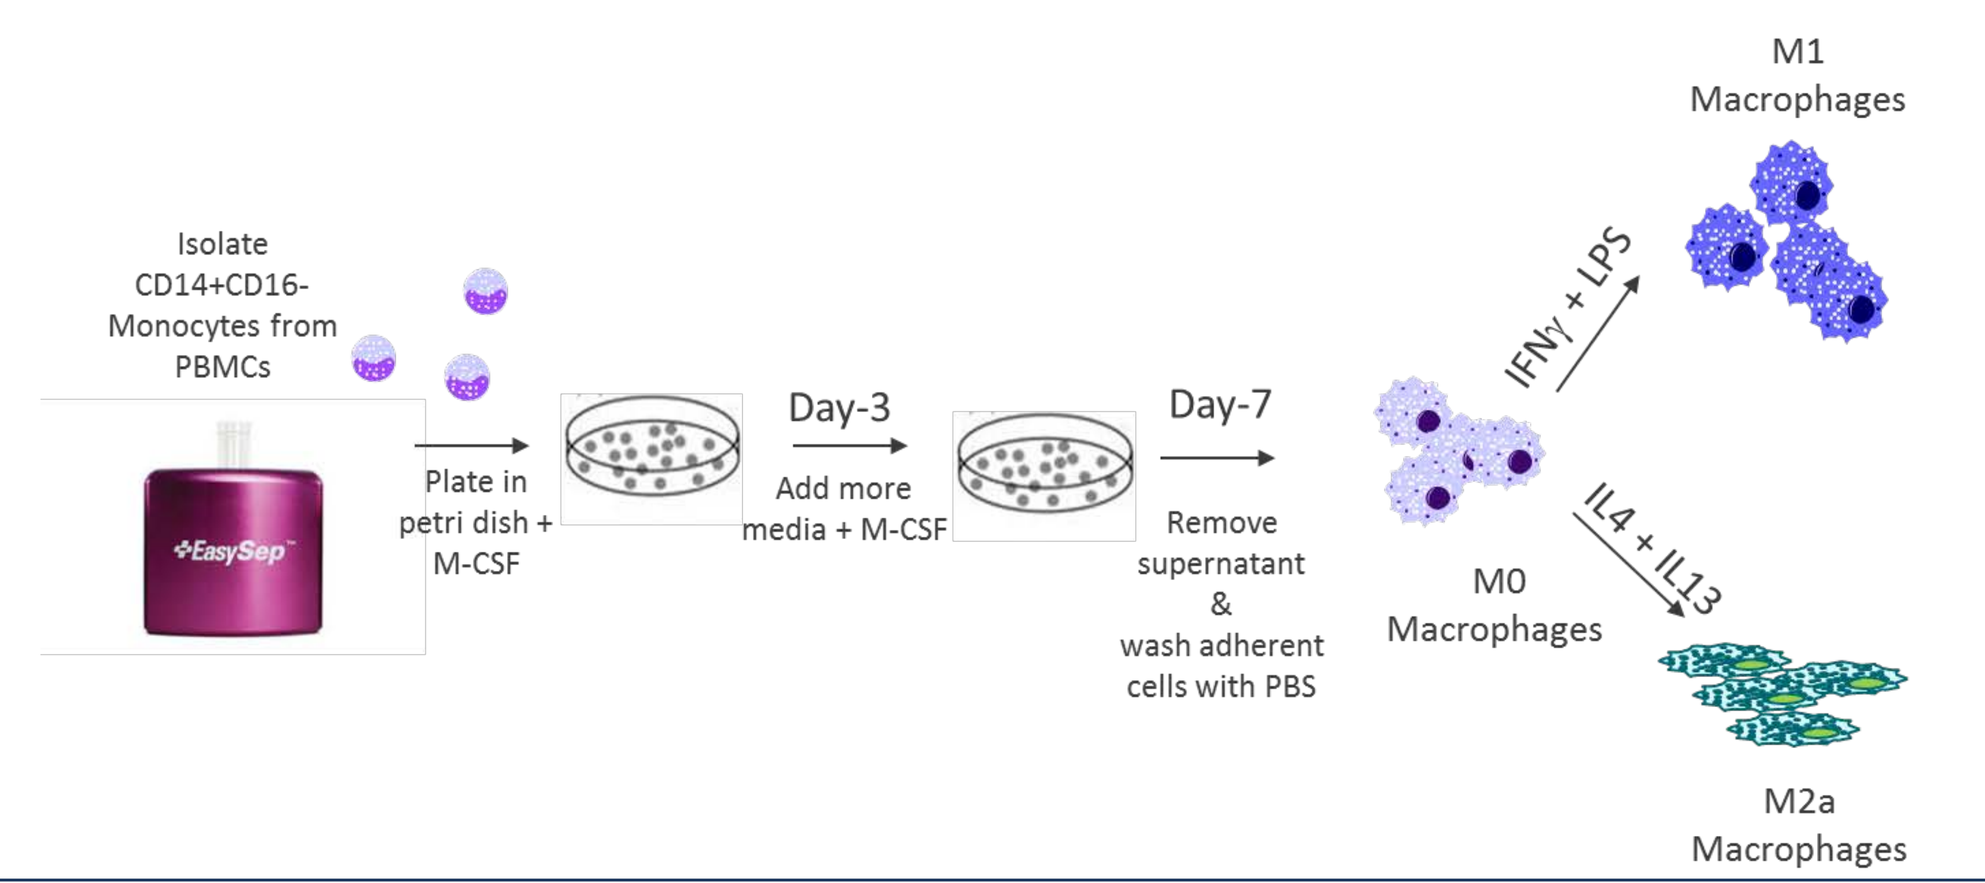
Isolate CD14+CD16- Monocytes from PBMCs
Plate in petri dish + M-CSF
Day-3
Add more media + M-CSF
Day-7
Remove supernatant & wash adherent cells with PBS
M0 Macrophages
IFNγ + LPS
M1 Macrophages
IL4 + IL13
M2a Macrophages

Figure 4. Generation of Human Macrophages from THP-1 Cell Line

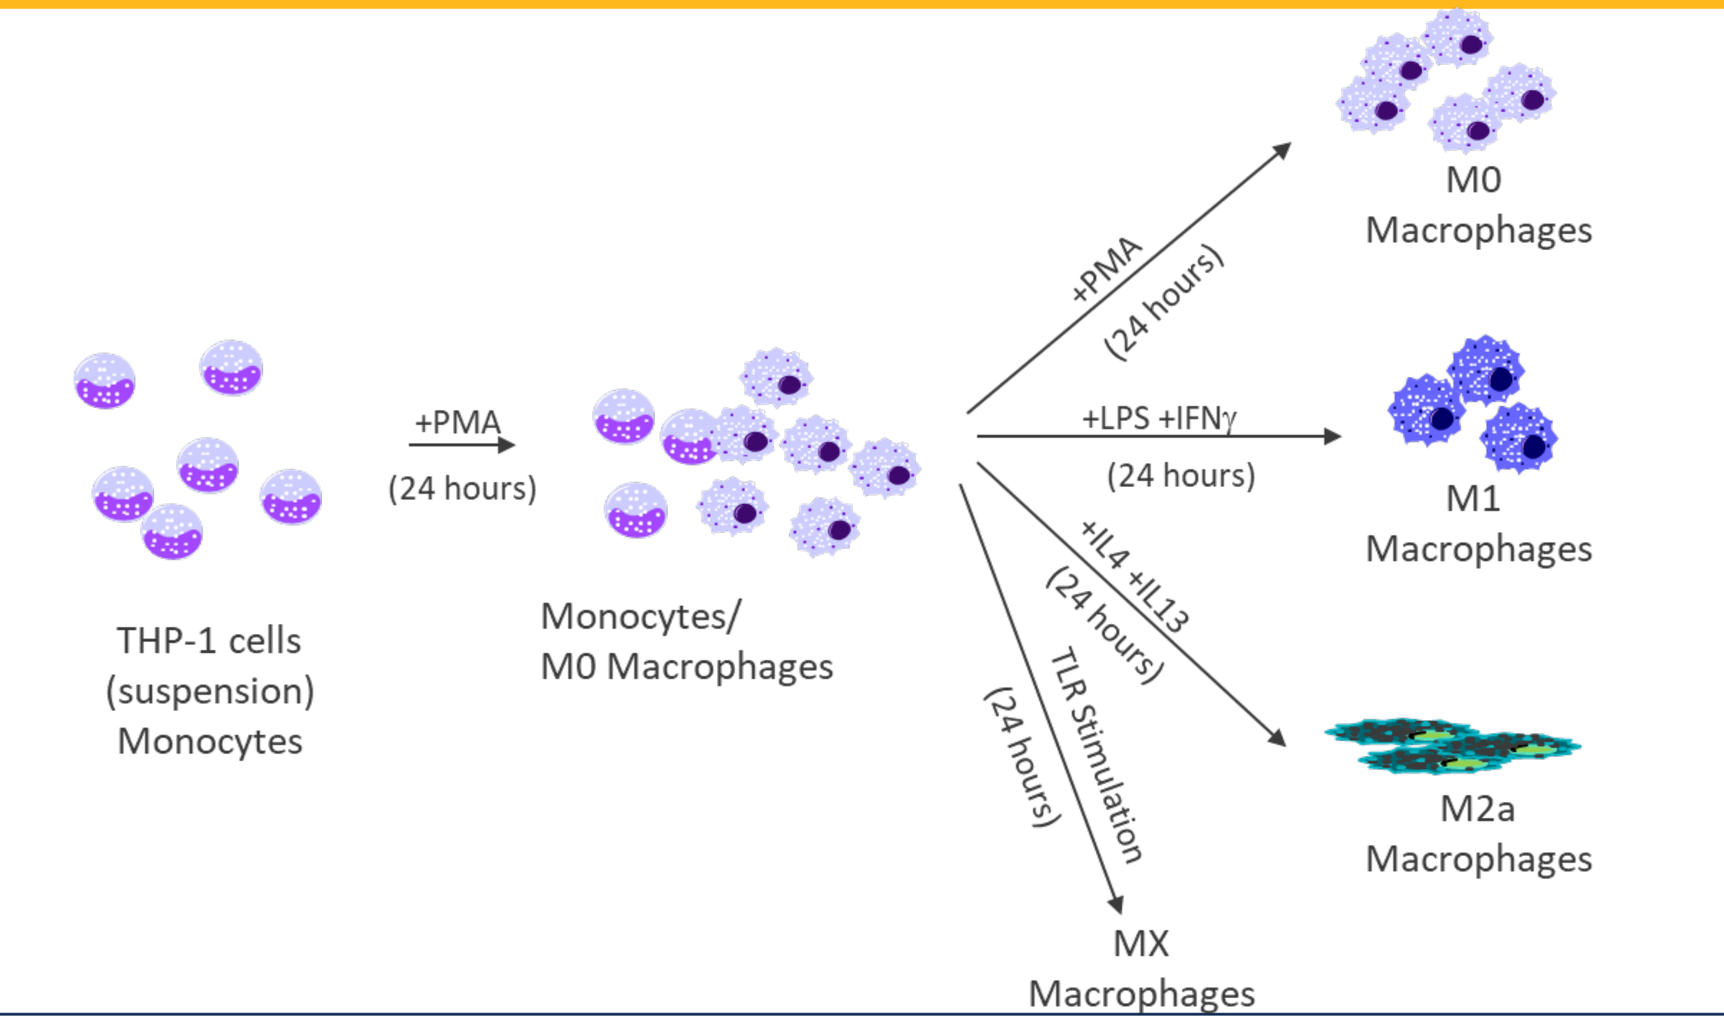
THP-1 cells (suspension) Monocytes
+PMA
(24 hours)
Monocytes/ M0 Macrophages
+PMA (24 hours)
M0 Macrophages
+LPS +IFNγ
(24 hours)
M1 Macrophages
+IL4 +IL13 (24 hours)
M2a Macrophages
TLR Stimulation (24 hours)
MX Macrophages

Figure 5. Generation of Human Primary Dendritic Cells

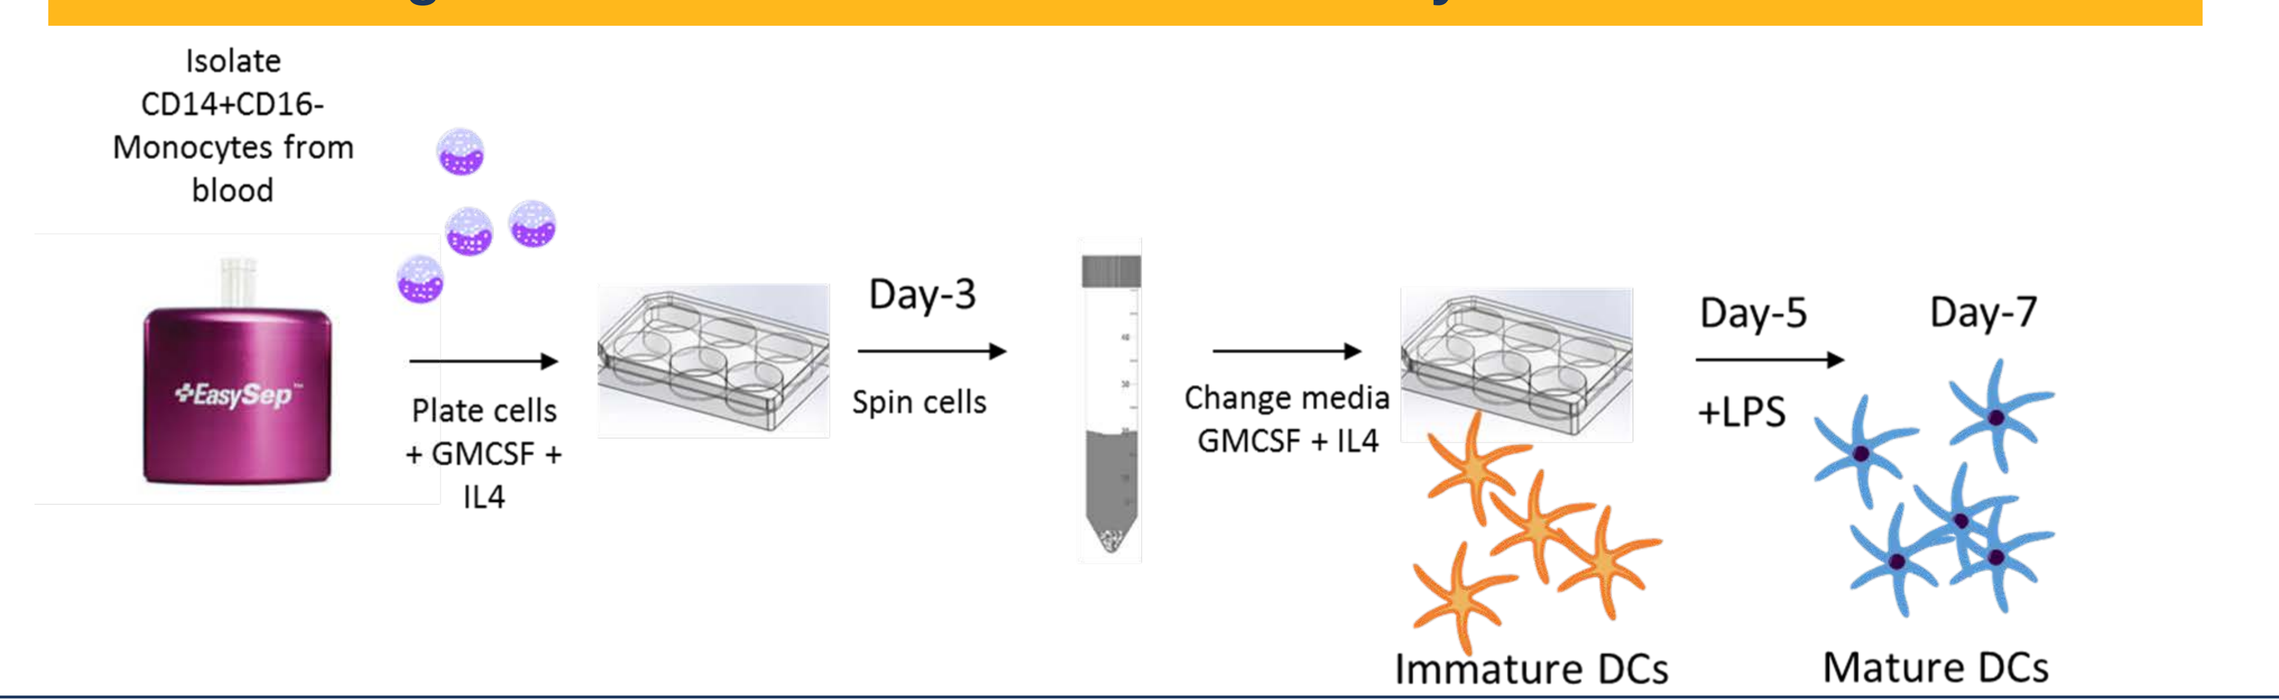
Isolate CD14+CD16- Monocytes from blood
Plate cells + GMCSF + IL4
Day-3
Spin cells
Change media GMCSF + IL4
Day-5
+LPS
Day-7
Immature DCs
Mature DCs

Figure 6. Generation of Primary Human Inducible T Regulatory Cells (iTregs)

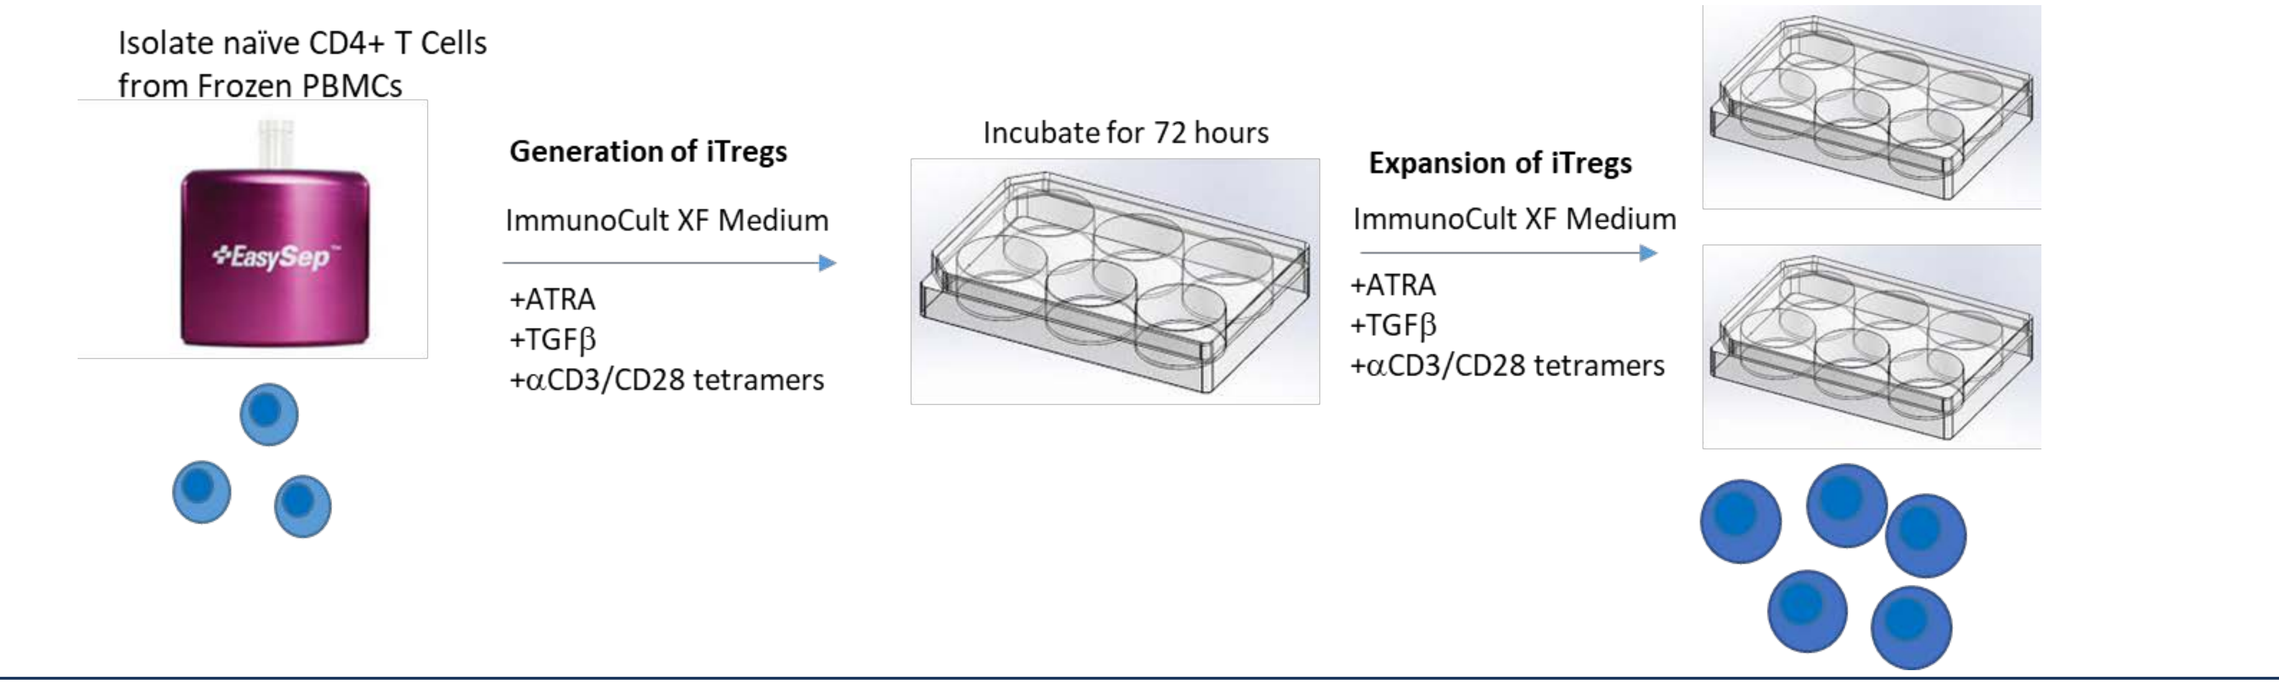
Isolate naïve CD4+ T Cells from Frozen PBMCs
Generation of iTregs
ImmunoCult XF Medium
+ATRA
+TGFβ
+αCD3/CD28 tetramers
Incubate for 72 hours
Expansion of iTregs
ImmunoCult XF Medium
+ATRA
+TGFβ
+αCD3/CD28 tetramers

Figures 3-6. Schematic representation showing the experimental procedures to generate: human primary macrophages (Figure 3); human macrophages from THP1 cell line (Figure 4); human primary dendritic cells (Figure 5) and primary human inducible T regulatory cells (Figure 6). NRP2 levels were measured using flow cytometry and were analyzed using FlowJo and statistical analysis performed using Prism.

In vivo Experimental Procedures

- Body weights (BW) were recorded and animals were anesthetized at 2-4% Isoflurane (1L/min) and dosed OP (oropharyngeally) with 10 ug LPS in 50 µL PBS or left untreated OR animals were administered IP (intraperitoneal) for systemic exposure to LPS.
- 24 hrs post LPS, mice were euthanized with lethal ketamine/xylazine cocktail (~300/30 mg/kg).

<u>**Acute Lung LPS Study:**</u>

- The lung and trachea were exposed & perfused for BALF (Bronchoalveolar lavage fluid) collection using 0.8 mL PBS through cannulated trachea.
- BALF cells were collected by centrifugation. RBC lysis was applied to BALF calls and stained for flow cytometry analysis.
- Lungs were perfused with MACS issue storage buffer, collected and trimmed of other tissues. , placed in storage buffer and kept at 4°C until dissociation to single cell suspension using. Miltenyi's lung dissociation kit and stained for flow cytometry analysis.

<u>**Systemic LPS Study:**</u>

- The spleen of animals were collected and dissociated using PBS and mechanical dissociation and stained for flow cytometry analysis.

Results

Figure 7. NRP2 Expression in Sarcoid Samples

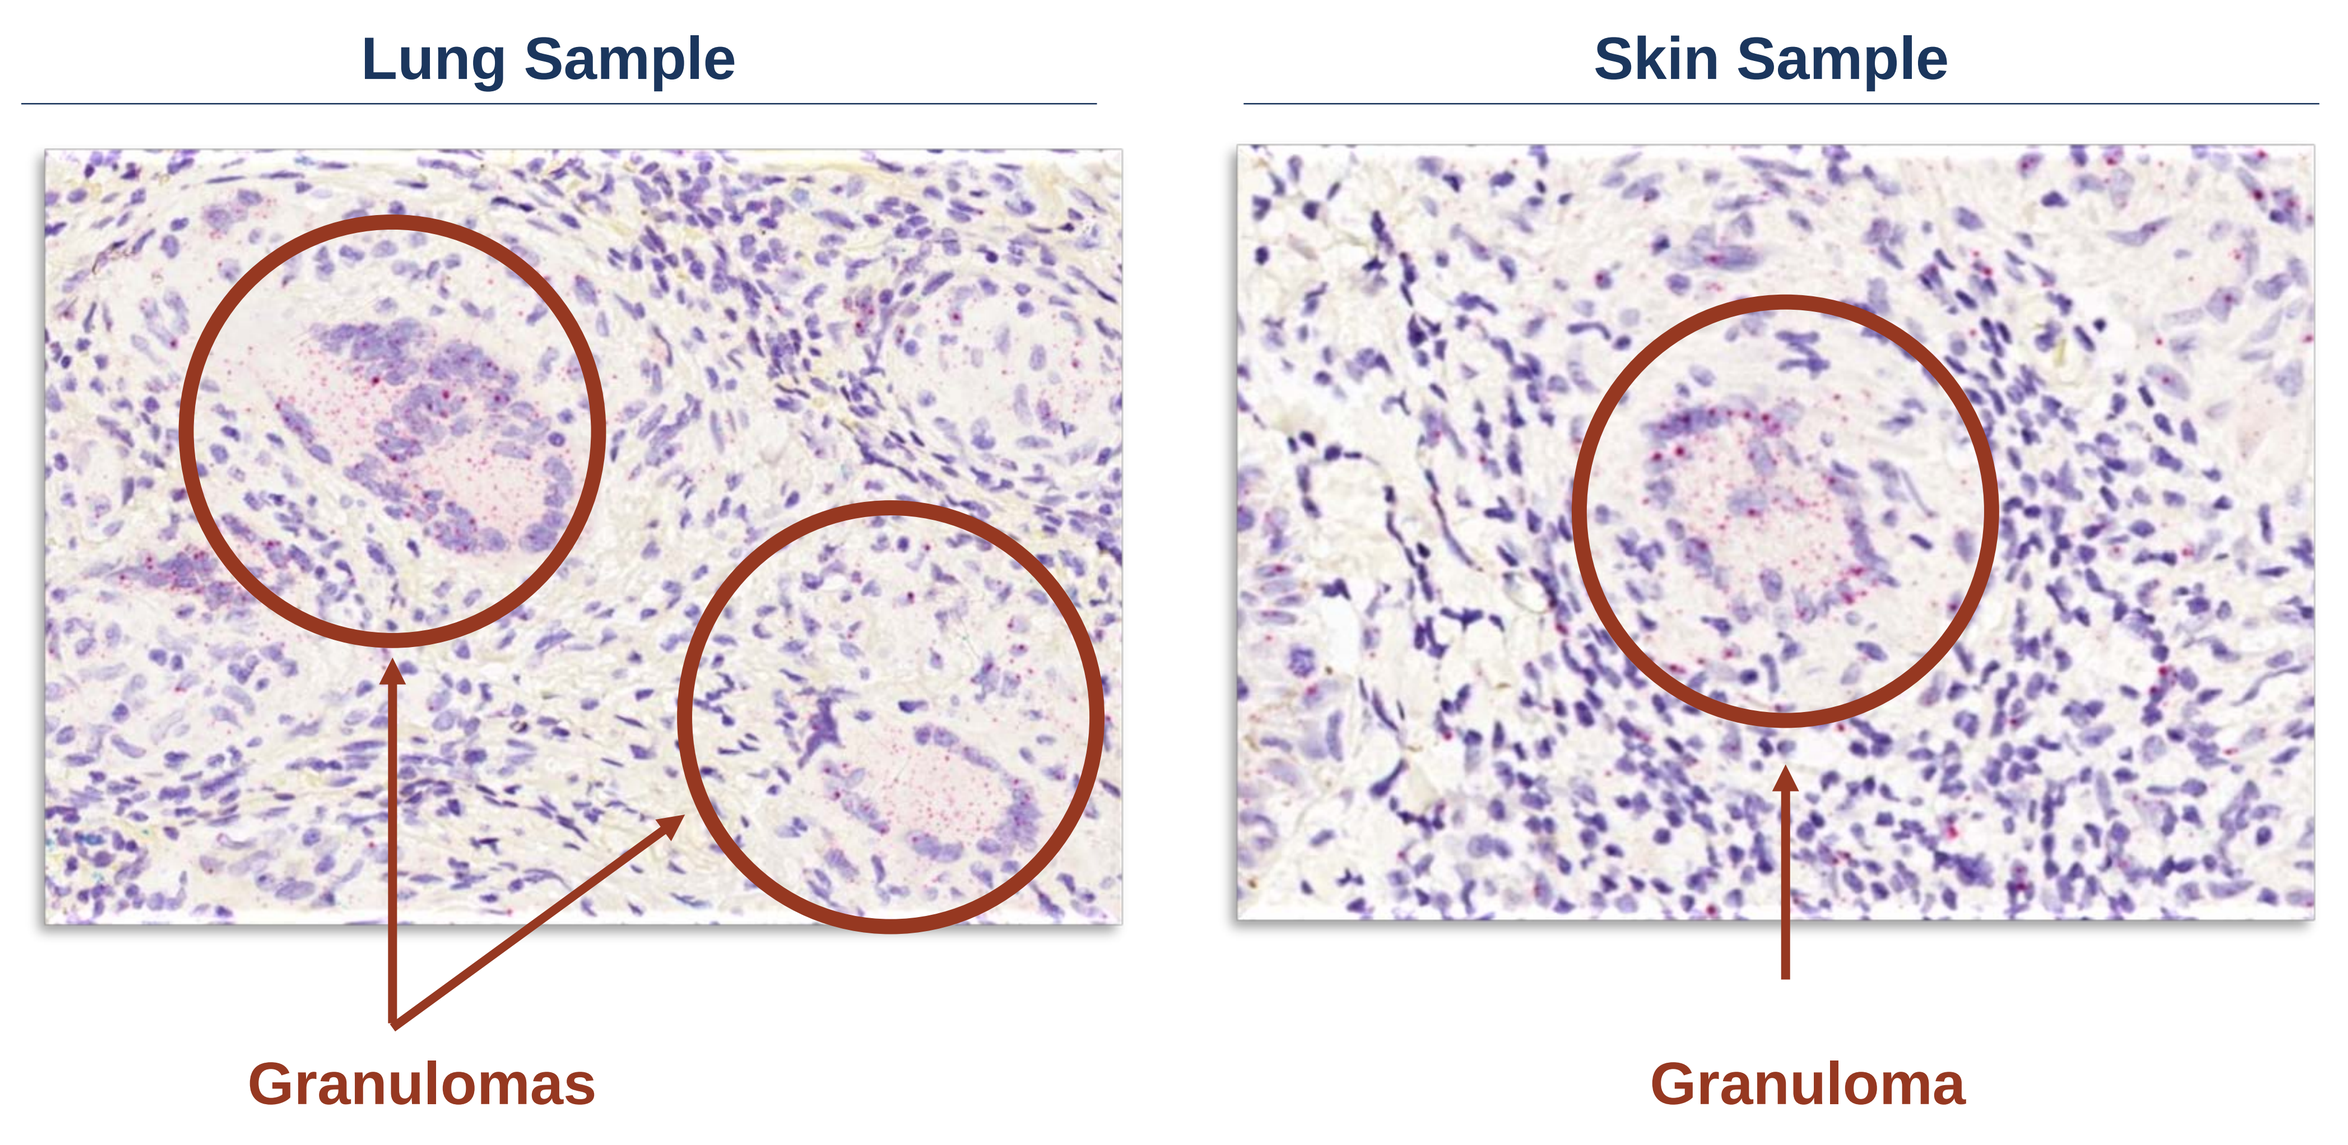
Lung Sample
Skin Sample
Granulomas
Granuloma

Figure 7. NRP2 Expression in Granuloma from Sarcoid A) Lung and B) Skin biopsies. Representative image of NRP2 ISH staining (red dots) in lung granulomas (A) and skin granulomas (B). A total of four lung samples and four skin samples were analyzed. NRP2 expression was low to moderate levels across the sample with high expression in granulomas from either lung or skin samples.

Figure 8. NRP2 Expression on Primary Macrophages

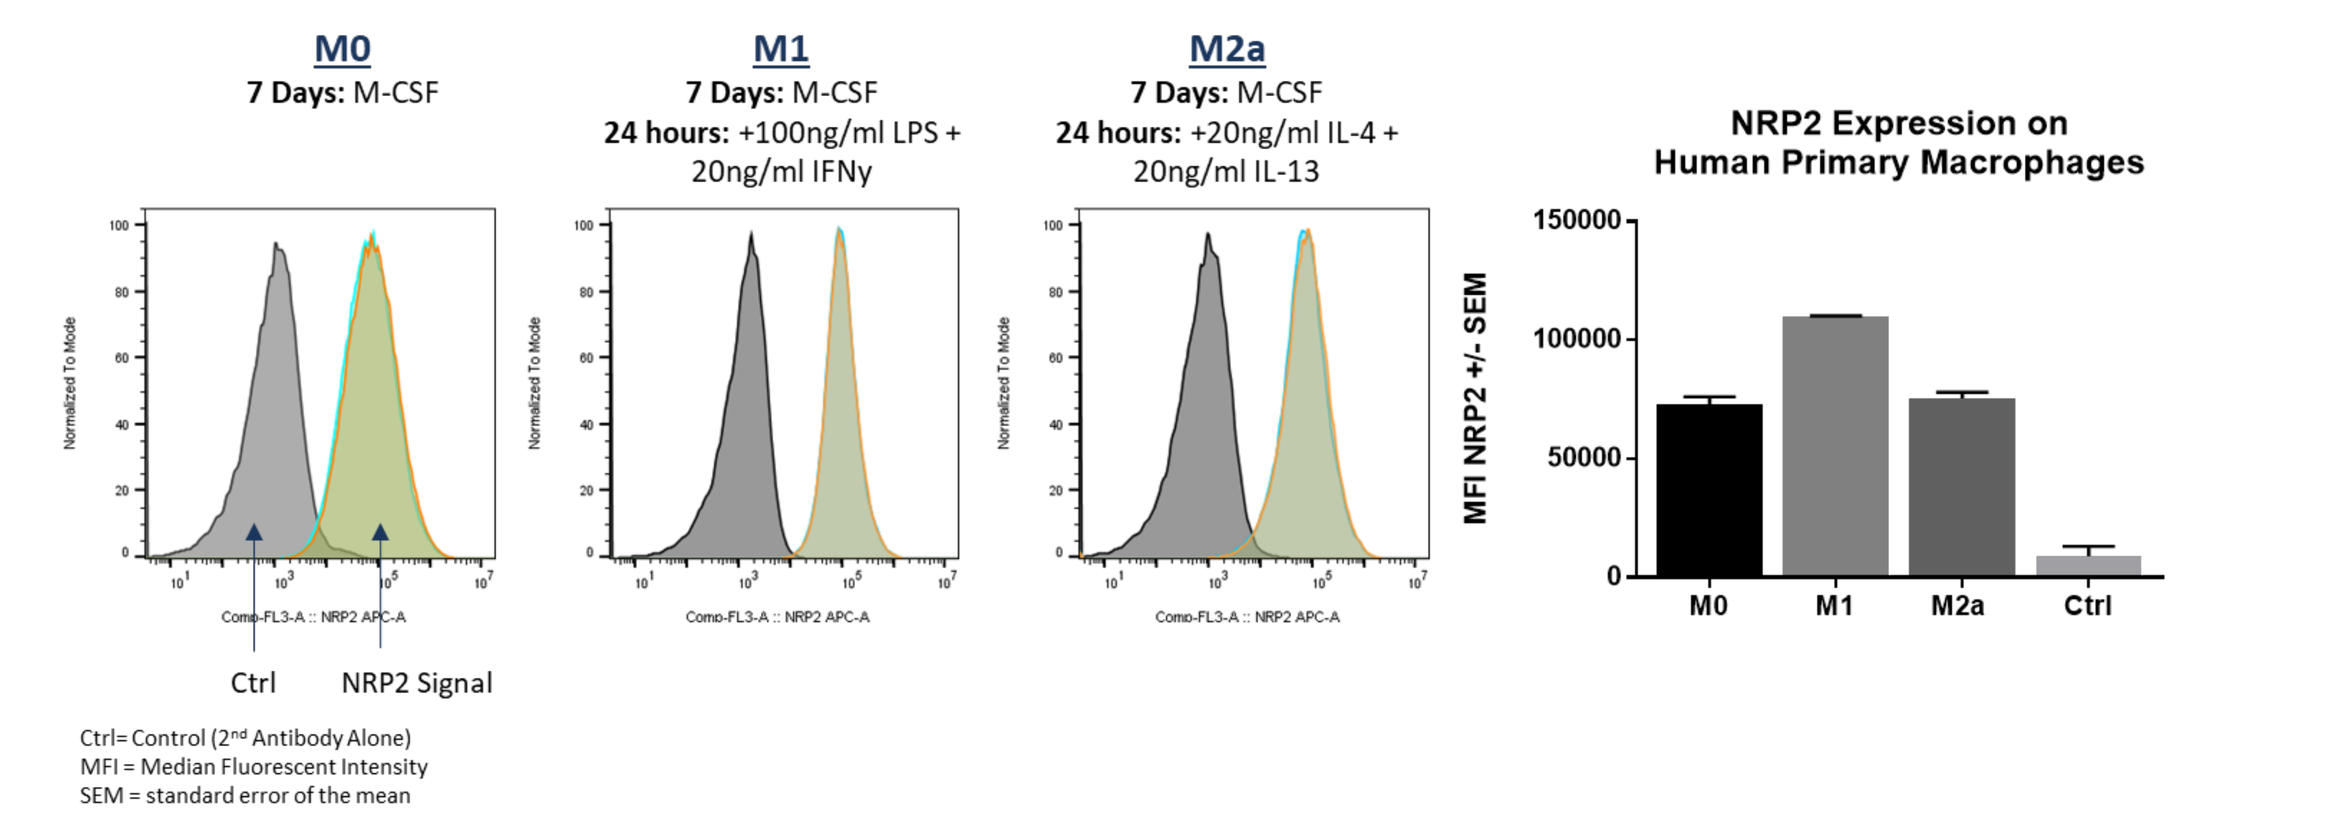
M0
7 Days: M-CSF
M1
7 Days: M-CSF
24 hours: +100ng/ml LPS + 20ng/ml IFNy
M2a
7 Days: M-CSF
24 hours: +20ng/ml IL-4 + 20ng/ml IL-13
Ctrl
NRP2 Signal
Ctrl= Control (2nd Antibody Alone)
MFI = Median Fluorescent Intensity
SEM = standard error of the mean
NRP2 Expression on Human Primary Macrophages
MFI NRP2 +/- SEM
M0
M1
M2a
Ctrl

Figure 8. Histogram representation of NRP2 expression detected on the cell surface of various in vitro generated primary macrophages cells (M0, M1, M2a) generated from CD14+CD16- monocytes. Bar graph representation of NRP2 mean fluorescent intensity (MFI) mean values +/- SEM.

Figure 9. NRP2 Expression on Human THP-1 Cell Line

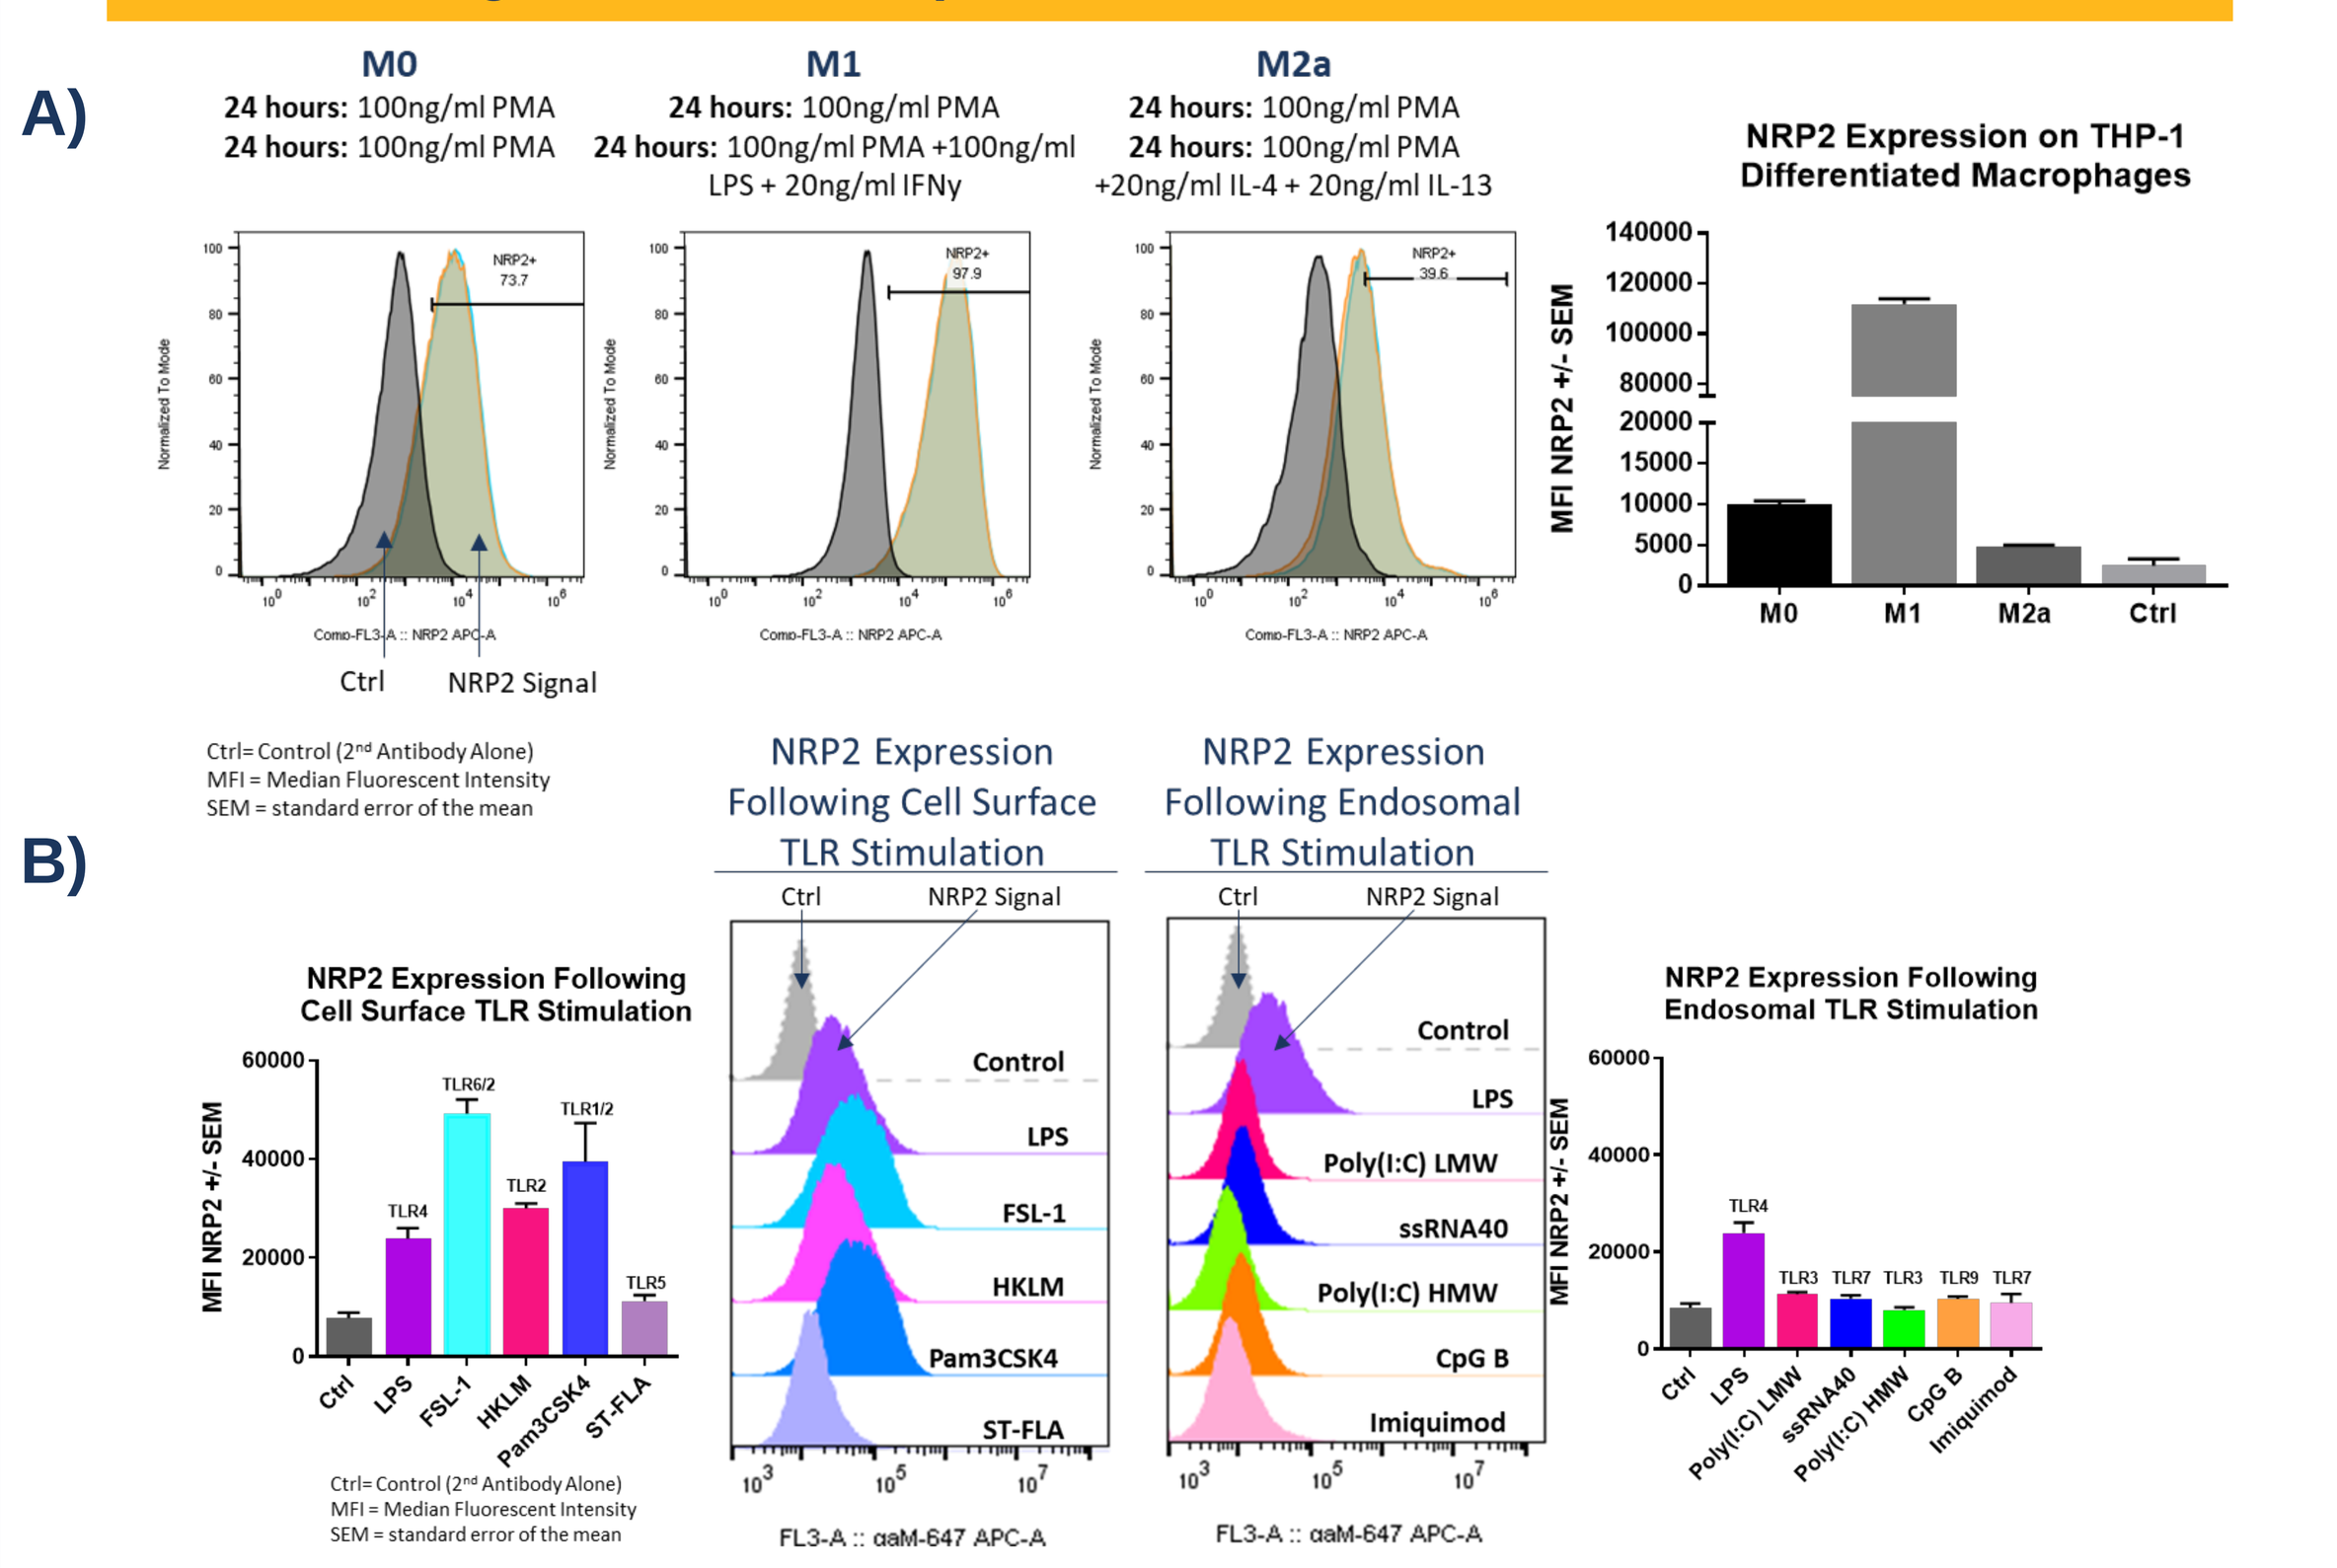
A)
M0
24 hours: 100ng/ml PMA
24 hours: 100ng/ml PMA
M1
24 hours: 100ng/ml PMA
24 hours: 100ng/ml PMA +100ng/ml LPS + 20ng/ml IFNy
M2a
24 hours: 100ng/ml PMA
24 hours: 100ng/ml PMA +20ng/ml IL-4 + 20ng/ml IL-13
NRP2 Expression on THP-1 Differentiated Macrophages
MFI NRP2 +/- SEM
Ctrl
NRP2 Signal
Ctrl= Control (2nd Antibody Alone)
MFI = Median Fluorescent Intensity
SEM = standard error of the mean
B)
NRP2 Expression Following Cell Surface TLR Stimulation
NRP2 Expression Following Endosomal TLR Stimulation
Control
LPS
FSL-1
HKLM
Pam3CSK4
ST-FLA
Poly(I:C) LMW
ssRNA40
Poly(I:C) HMW
CpG B
Imiquimod

Figure 9. A) Histogram representation of NRP2 expression detected on the cell surface of various in vitro generated myeloid cells (M0, M1, M2a) from THP-1 Cell line and Bar graph representation of NRP2 MFI mean values +/- SEM. **B)** Histogram representation of NRP2 expression detected on the cell surface of M0 THP-1 derived cells stimulated with various TLR ligands as indicated and bar graph of NRP2 MFI mean values +/- SEM.

Figure 10. NRP2 Expression on Human Primary Dendritic Cells

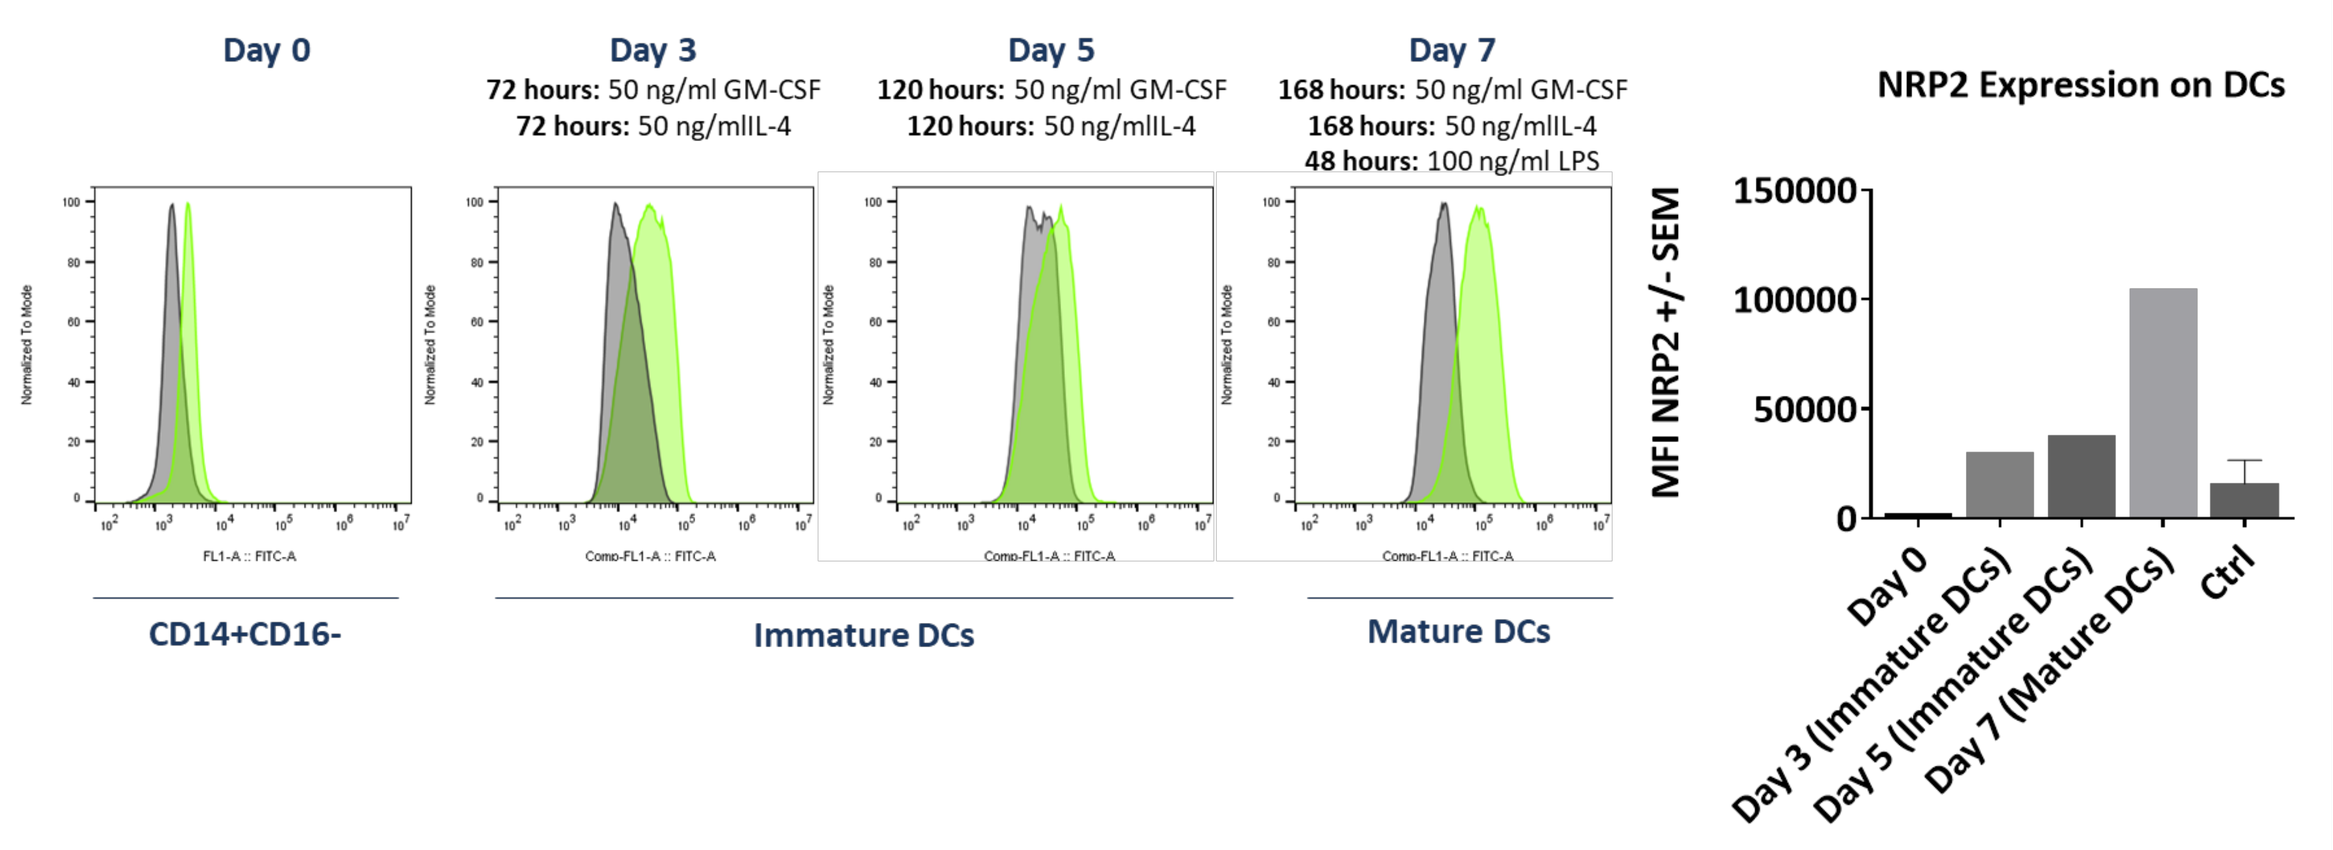
Day 0
Day 3
72 hours: 50 ng/ml GM-CSF
72 hours: 50 ng/mlIL-4
Day 5
120 hours: 50 ng/ml GM-CSF
120 hours: 50 ng/mlIL-4
Day 7
168 hours: 50 ng/ml GM-CSF
168 hours: 50 ng/mlIL-4
48 hours: 100 ng/ml LPS
CD14+CD16-
Immature DCs
Mature DCs
NRP2 Expression on DCs
MFI NRP2 +/- SEM

Figure 10. Histogram representation of NRP2 expression detected on the cell surface of primary human DCs at Day 0, 3, 5 and 7 generated from CD14+CD16- monocytes. Bar graph representation of NRP2 MFI mean values +/- SEM.

Figure 11. NRP2 Expression on Human Inducible Tregs

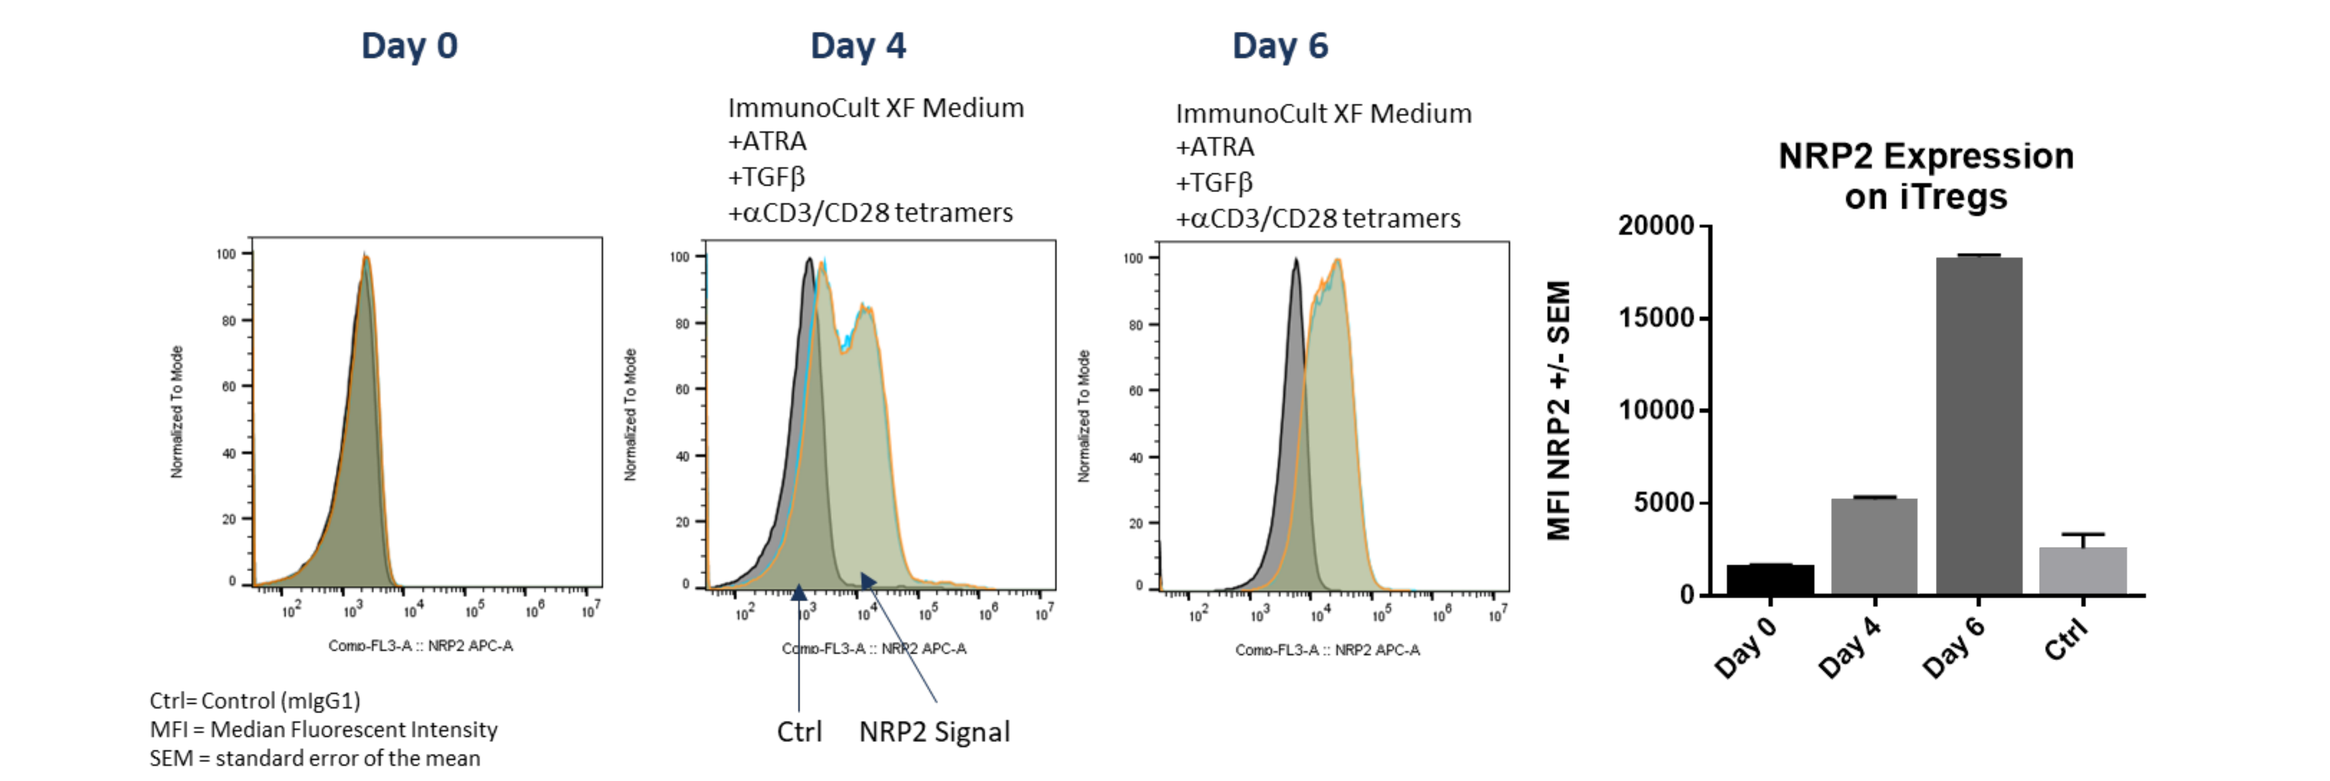
Day 0
Day 4
Day 6
ImmunoCult XF Medium
+ATRA
+TGFβ
+αCD3/CD28 tetramers
NRP2 Expression on iTregs
MFI NRP2 +/- SEM
Ctrl= Control (mIgG1)
MFI = Median Fluorescent Intensity
SEM = standard error of the mean
Ctrl
NRP2 Signal

Figure 11. Histogram representation of NRP2 expression detected on the cell surface of primary human inducible Tregs on Day 0, 4 and 6 generated from CD4+ naïve T cells. Bar graph representation of NRP2 MFI mean values +/- SEM.

Figure 12. *in vivo* NRP2 Expression

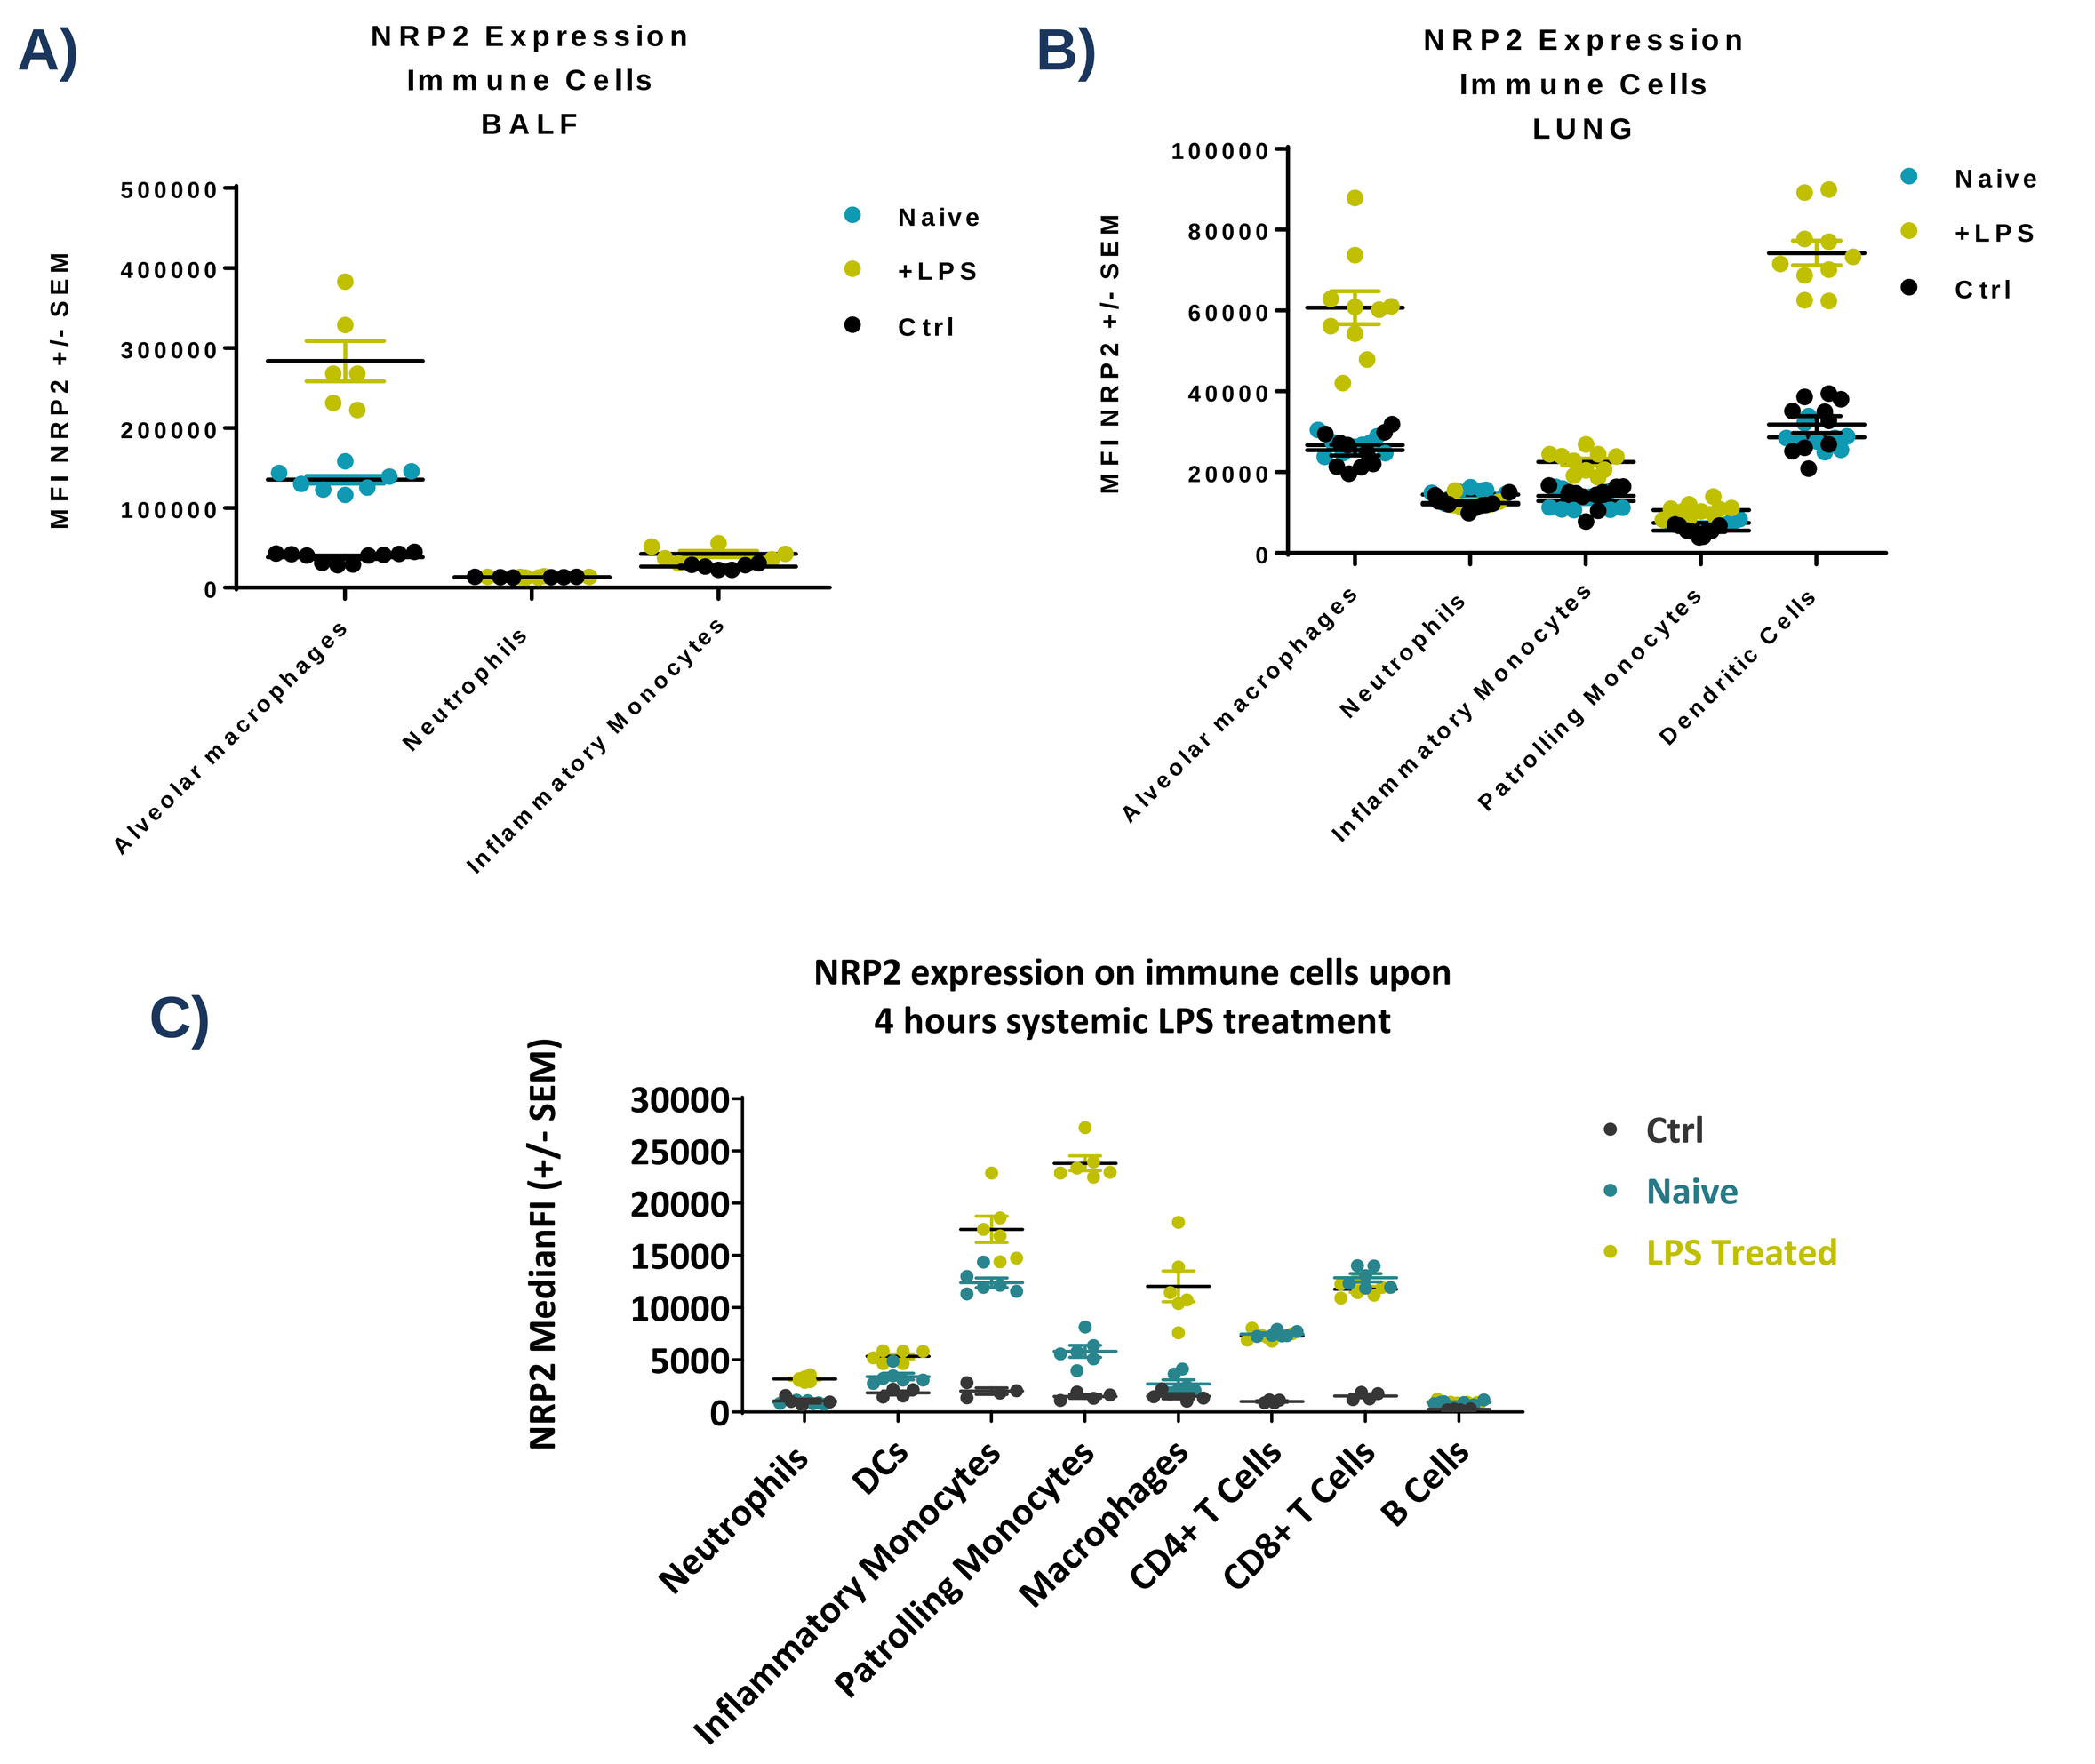
A)
NRP2 Expression Immune Cells BALF
MFI NRP2 +/- SEM
Naive
+LPS
Ctrl
Alveolar macrophages
Neutrophils
Inflammatory Monocytes
B)
NRP2 Expression Immune Cells LUNG
Patrolling Monocytes
Dendritic Cells
C)
NRP2 expression on immune cells upon 4 hours systemic LPS treatment
NRP2 MedianFI (+/- SEM)
Ctrl
Naive
LPS Treated
Neutrophils
DCs
Inflammatory Monocytes
Patrolling Monocytes
Macrophages
CD4+ T Cells
CD8+ T Cells
B Cells

Figure 12. Scattered plot of NRP2 cell surface expression detected on the cell surface of immune cells in **A)** BALF (alveolar macrophages, neutrophils & inflammatory monocytes), **B)** lung tissue, (alveolar macrophages, neutrophils, inflammatory monocytes, patrolling monocytes and dendritic cells) **C)** spleen (B cells, neutrophils, dendritic cells, CD4+ T cells, CD11b+ myeloid cells, inflammatory monocytes and patrolling monocytes) of animals challenged with LPS. Mean values +/- SEM.

Conclusions

- NRP2 was identified as specific binding partner for ATYR1923 (Paz et al. Keystone Poster 2019; Xu et al. ATS Poster #P1173)
- NRP2 was expressed within the granulomas of both lung and skin biopsies obtained from sarcoidosis patients, predominantly localized within the inner structure of the granuloma where Langham's giant cells (myeloid origin) reside
- NRP2 was highly expressed on macrophages, dendritic cells and Tregs, which are all cell types involved in granuloma formation
- NRP2 was highly expressed following immunostimulation (TLR agonists) in both *in vitro* and *in vivo* systems
- We clearly show that NRP2 expression can be detected on key immune cells known to play an important role in inflammation and granuloma formation. These findings highlight the potential of ATYR1923 to exert its effect on various immune cells directly related to the pathology of the target patient population.

References

1. Paz S, Polizzi C, Chu D, Burkart C, King D, Ogilvie K, Rosengren S. ATYR1923 Reduces Neutrophil Infiltration in an Acute Lipopolysaccharide (LPS) Lung injury Model. 2019 *Keystone Symposia Conference. B7: Myeloid Cells*, Santa Fe, NM. Poster Link
2. Xu Z, Chong Y, Crampton S, Adams R, Rauch K, Paz S, Chu D, Zhai L, Burman L, Ampudia J, Rosengren S, King D, Nangle L. ATYR1923 Specifically Binds to Neureopilin-2, a Novel Therapeutic Target for the Treatment of Immune-Mediated Diseases. *ATS 2020.* Abstract Link
3. Broos, EB, van Nimwegen M, Hoogsteden HC, Hendriks RW, Kool M, ven der Blink B. Granuloma formation in pulmonary sarcoidosis. *Frontiers in Immunology* **4,** 437 (2013). https://www.frontiersin.org/article/10.3389/fimmu.2013.00437
4. Inaoka, P.T., Shono, M., Kamada, M. *et al.* Host-microbe interactions in the pathogenesis and clinical course of sarcoidosis. *J Biomed Sci* **26,** 45 (2019). https://doi.org/10.1186/s12929-019-0537-6
5. Miedema, J.R., Kaiser, Y.K., Broos, C.E., Wijsenbeek, M.S., Grunewald, J., Kool, M. Th17-lineage cells in pulmonary sarcoidosis and Lofgren's syndrome: Friend or foe?. *J of Autoimmunity* **87, 82-89** (2018). https://doi.org/10.1016/j.jaut.2017.12.012