A Monoclonal Antibody with Tissue Factor Pathway Inhibitor (TFPI) Neutralizing Activity Improves the Coagulation Parameters of Hemostatic Assays Performed with Hemophilic Whole Blood

International Society on Thrombosis and Haemostasis
2017 Congress
Berlin, Germany
Abstract # PB 1124

Authors:

Sunita Patel-Hett [1]
Erika J. Martin [2]
Pengling Sun [1]
Swapnil Rakhe [1]
Debra D. Pittman [1]
Melinda Nolte [2]
Janice Kuhn [2]
J. Christian Barrett [2]
Donald Brophy [2]
John E. Murphy [1]

Affiliations:

[1] Hematology Division
Rare Disease
Research Unit
Pfizer, Inc
Cambridge, Massachusetts
USA

[2] Virginia
Commonwealth
University
Richmond, Virginia
USA

Acknowledgments:

The authors acknowledge all individuals at Pfizer, Inc. who contributed to the research and development of PF-06741086. The authors also acknowledge the hemophilia volunteers who participated in this study.

Please scan this QR code with your smartphone app to view an electronic version of this poster. If you do not have access to a smartphone, please access the poster via the following link: http://congressdownload.pfizer.com/isth_2017_international_society_on_thrombosis_and_haemostasis_308_benefix_sunita_patel_hett_1124.html

Introduction

TFPI is an endogenous inhibitor of Factor Xa (FXa) and Factor VIIa (FVIIa), both critical to thrombin generation during coagulation. Restoring FXa and FVIIa activities by inhibiting TFPI may enhance coagulation in hemophilia, where deficiencies in Factor VIII (FVIII) or Factor IX (FIX) impair thrombin generation. Physiologically relevant pools of TFPI affect coagulation exist free in plasma and are released from platelets upon activation.

We investigated the effect of PF-06741086, a TFPI neutralizing human monoclonal antibody, in comparison to recombinant FVIII or FIX in hemostatic assays performed with whole blood from hemophilic patients with and without inhibitors. We also investigated the baseline variability and reproducibility of PF-06741086 in restoring hemostasis by comparing its effect in whole blood collected from the same patients on differing days.

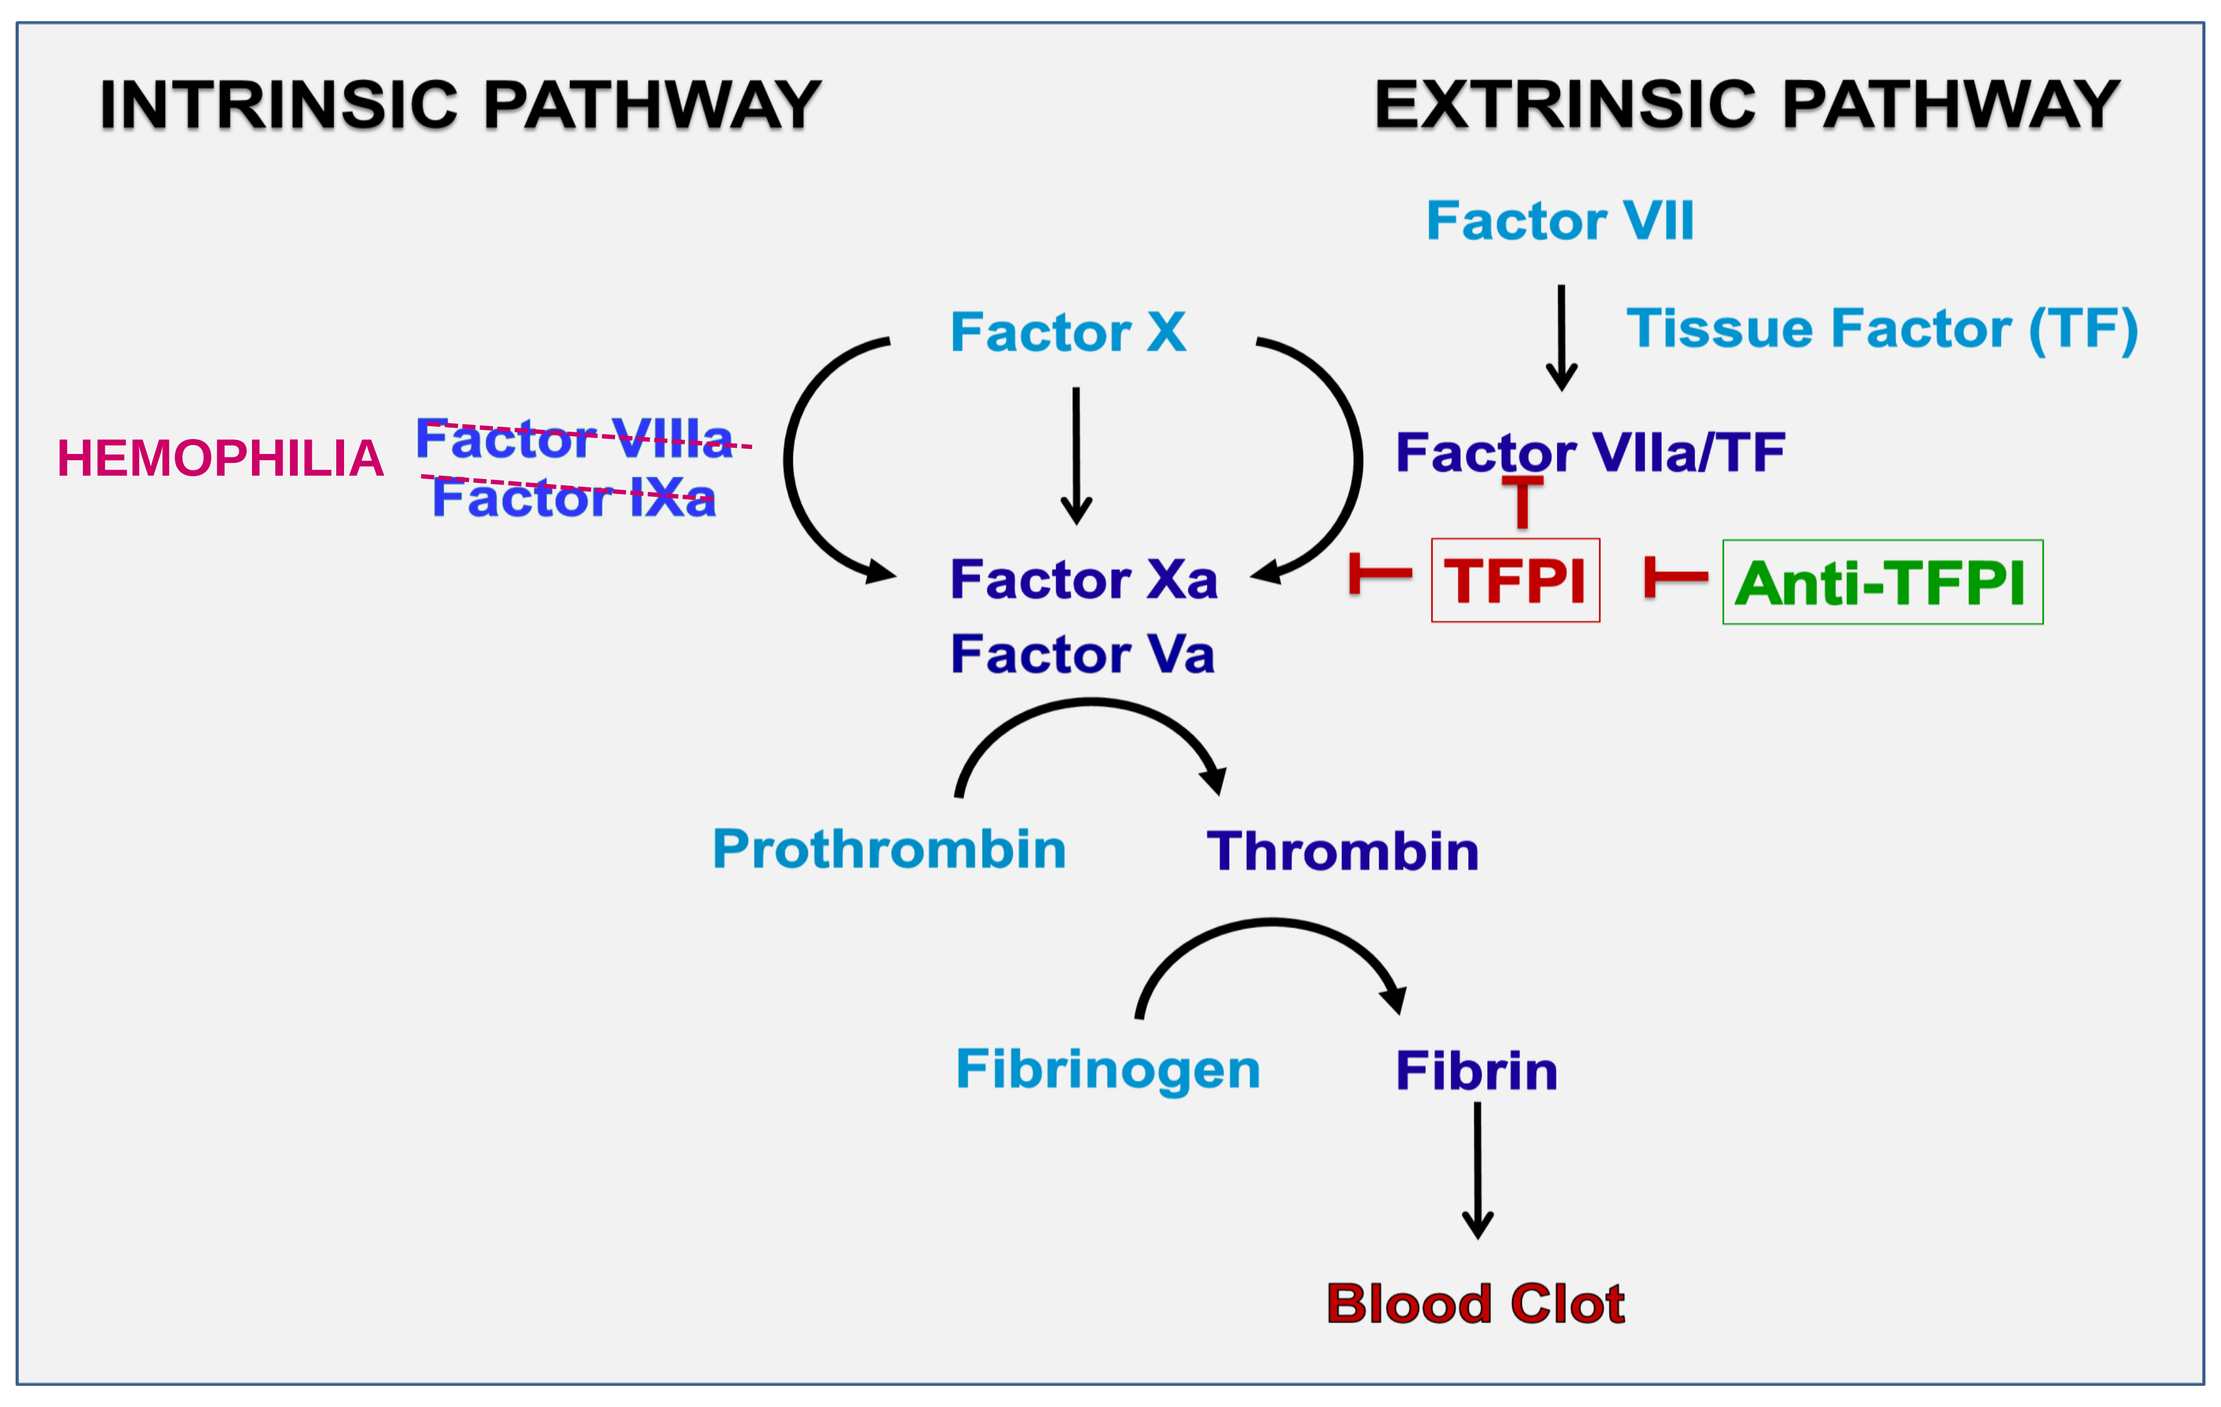
INTRINSIC PATHWAY
EXTRINSIC PATHWAY
Factor VII
Tissue Factor (TF)
Factor X
HEMOPHILIA
~~Factor VIIIa~~
~~Factor IXa~~
Factor VIIa/TF
Factor Xa
Factor Va
TFPI
Anti-TFPI
Prothrombin
Thrombin
Fibrinogen
Fibrin
Blood Clot

Figure 1. In hemophilia, deficiencies in FVIII or FIX prevent the formation of thrombin necessary for stable clot formation. TFPI is an endogenous plasma inhibitor of coagulation, that limits the activity of Factor Xa and Factor VIIa/Tissue Factor in the extrinsic pathway. Inhibition of TFPI by anti-TFPI may restore sufficient thrombin formation within the extrinsic pathway to allow for stable blood clot formation.

Methods

Whole blood was collected from hemophilia patient volunteers including severe Hemophilia A, hemophilia A with inhibitors and moderate Hemophilia B under an institutional review board approved protocol. Whole blood was collected by venipuncture into blood collection tubes containing 0.105ml/L trisodium citrate. Whole blood was dosed ex vivo with vehicle (20 mM HEPES, 150 mM NaCl, pH 7.4), PF-06741086 (Pfizer, Inc.), recombinant Factor VIII (rFVIII) (Pfizer, Inc.) or recombinant Factor IX (rFIX) (Pfizer, Inc.) and analyzed in coagulation assays. Recombinant factors were dosed into untreated whole blood to achieve either 5, 10 or 40% of normal activity following determination of plasma activity by the activated partial thromboplastin time assay. PF-06741086 was dosed ex vivo in the range of 20-100 nM to approximate anticipated steady state maximal plasma concentrations following in vivo dosing. For analysis of day to day variability in responses, blood was collected from individual donors on differing days whole blood clotting was measured.

Rotational thromboelastometry (ROTEM): Whole blood was analyzed for clotting by modified thromboelastometry using ROTEM Delta Hemostasis analyzers (Pentapharm GmbH, Munich, Germany) and Pentapharm software version 2.0.0. The device temperature was set to 37°C and the maximum run time set to 90 minutes. Whole blood was analyzed using a 1:200,000 final dilution of Dade Innovin lipidated tissue factor reagent (Siemens Healthcare Diagnostics Inc., Tarrytown, New York) and recalcification with 10 mM $CaCl_2$. ROTEM parameters were monitored and exported from Pentapharm software and analyzed using Microsoft Excel 2010 version or GraphPad Prism version 6.03. Values not measured by the software due to a lack of hemostasis were assigned 60 minutes for clotting time, 0 for angle and 0 for amplitude.

Thromboelastography (TEG): TEG analysis was performed using a TEG 5000 Thromboelstaograph Haemostasis Analyzer (Haemonetics, Braintree, MA, USA). The device temperature was set to 37°C and the maximum run time set to 120 minutes. Clotting was initiated using addition of 10 mM calcium chloride and a 1:200,000 final dilution of Dade Innovin lipidated tissue factor (Siemens Healthcare). TEG parameters were monitored and obtained from TEG software and were analyzed using Microsoft Excel 2010 version or GraphPad Prism version 6.03. Values not measured by the software due to a lack of hemostasis were assigned 60 minutes for clotting time, 0 for angle and 0 for amplitude.

Statistical Analyses: Responses of vehicle, PF-06741086, rFVIII or rFIX treated whole blood samples were compared following normalization of whole blood clotting parameters. For normalization, the percent change in response was calculated with respect to vehicle treatment. Log transformed fold changes from vehicle-treatment values were plotted for each treatment condition per day using SAS version 9.4 software (SAS Institute Inc., Cary, NC, USA).

Results

Figure 2. PF-06741086 normalizes hemostasis in severe FVIII deficient whole blood

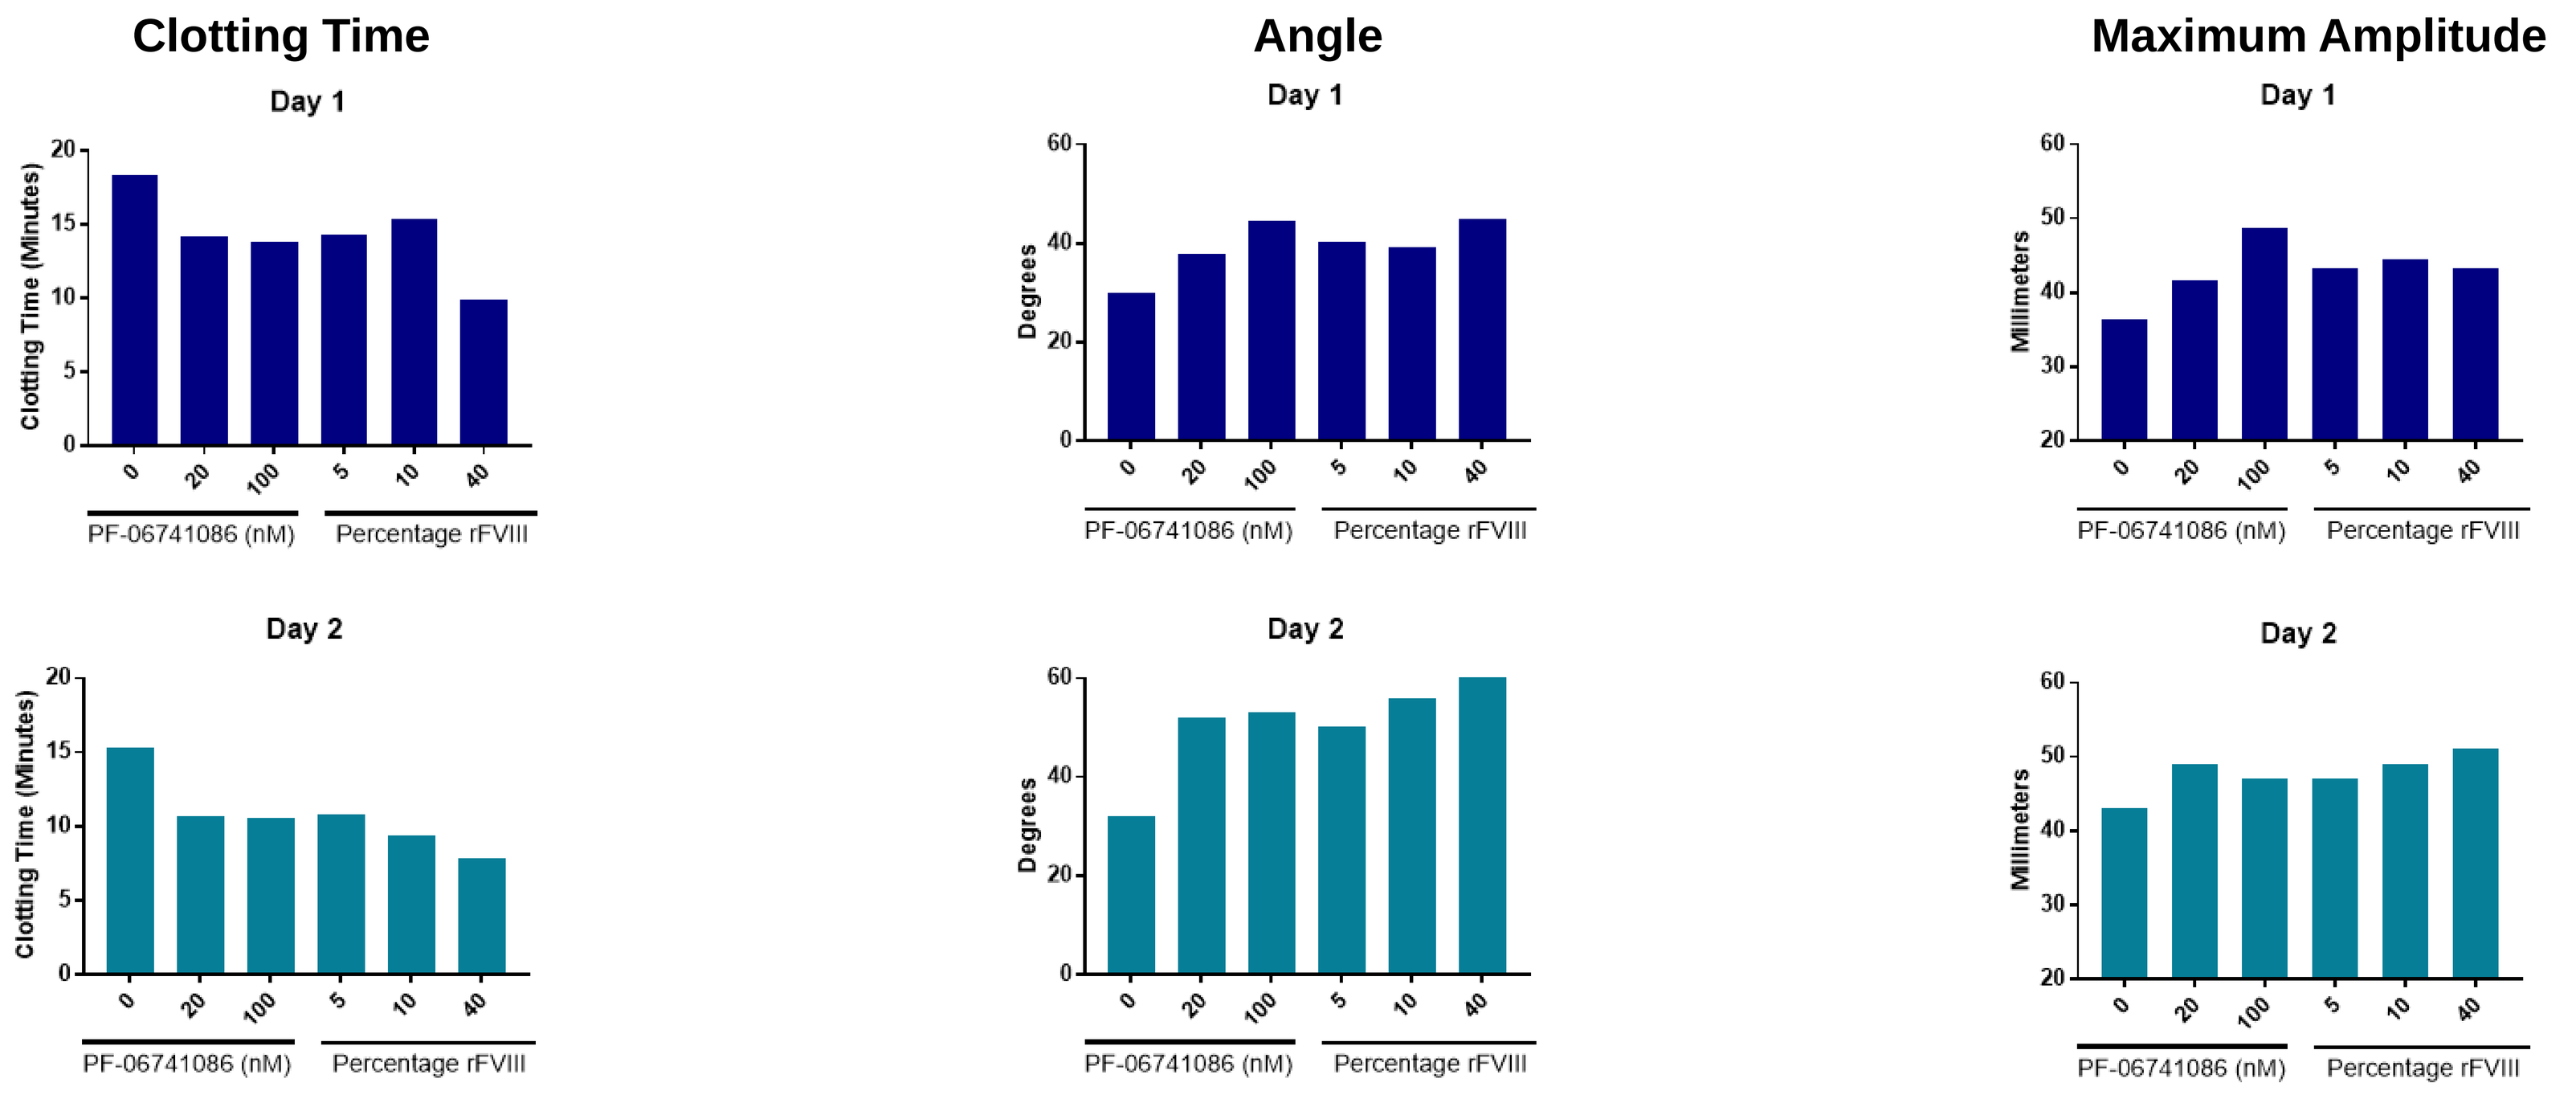
Clotting Time
Angle
Maximum Amplitude
Day 1
Day 2
Clotting Time (Minutes)
Degrees
Millimeters
PF-06741086 (nM)
Percentage rFVIII

Figure 2. Response of PF-06741086 in severe Factor VIII deficient whole blood
Ex vivo dosing of PF-06741086 to a final concentration of 20 nM or 100 nM in severe FVIII deficient whole blood shows a decrease in clotting time and increases in both angle and amplitude when analyzed by TEG (Day 1) or ROTEM (Day 2). Clotting times of PF-06741086 treated samples exhibit responses similar to those observed following additions of rFVIII.

Figure 3. PF-06741086 normalized hemostasis in severe FVIII deficient with inhibitor whole blood

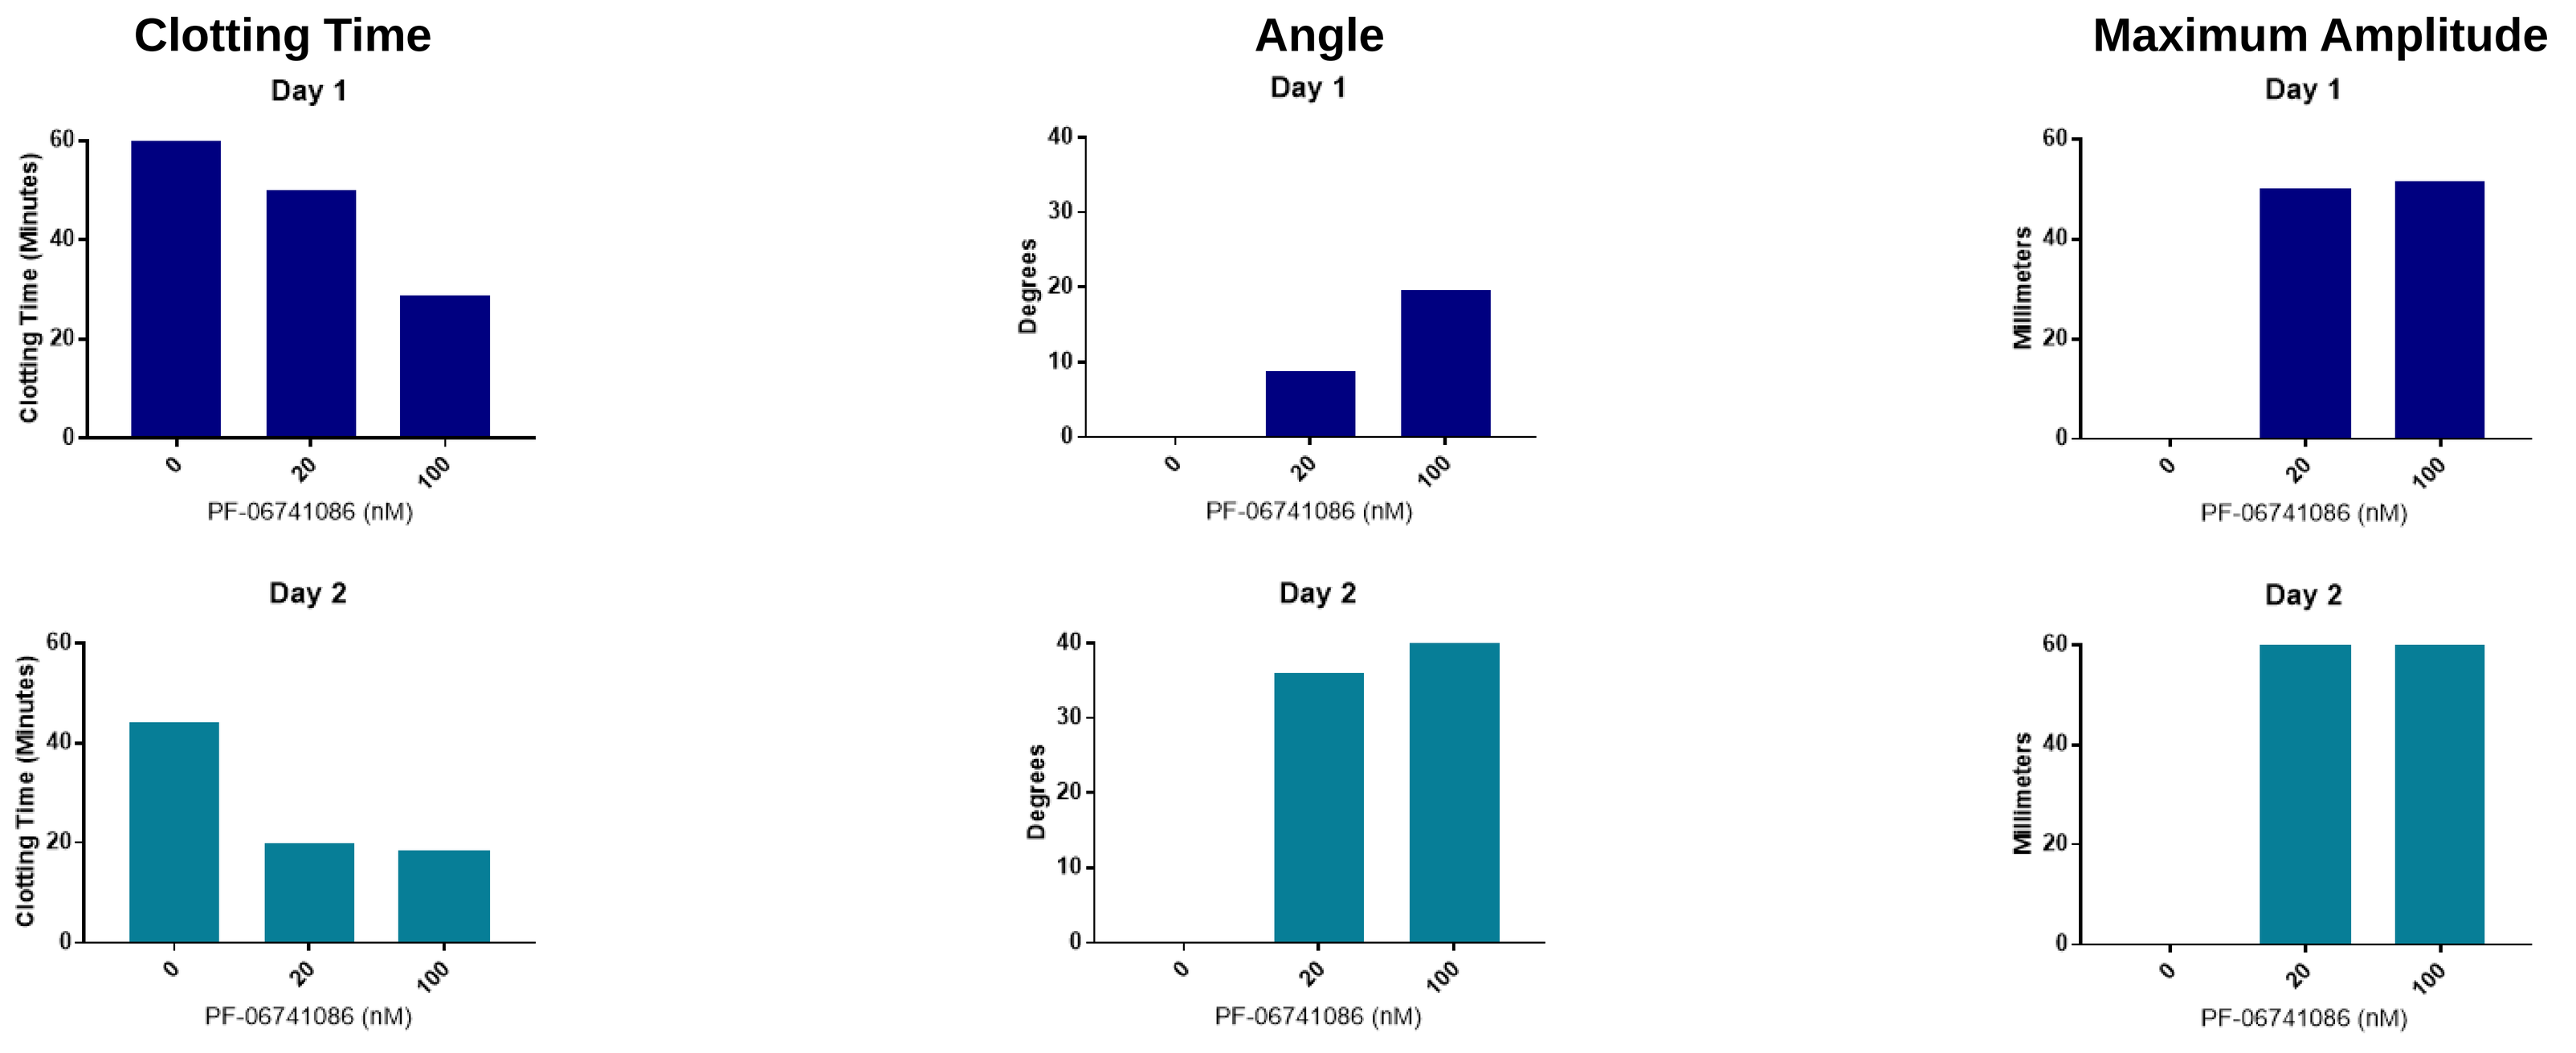
Clotting Time
Angle
Maximum Amplitude
Day 1
Day 2
Clotting Time (Minutes)
Degrees
Millimeters
PF-06741086 (nM)

Figure 3. Response of PF-06741086 in severe Factor VIII with FVIII inhibitor whole blood
Ex vivo dosing of PF-06741086 to a final concentration of 20 nM or 100 nM in severe Factor VIII deficient with inhibitor whole blood shows a dose-dependent decrease in clotting time, and increases in both angle and amplitude compared to vehicle when analyzed by TEG (Day 1) or ROTEM (Day 2). For whole blood samples that failed to clot, a clotting time of 60 minutes, angle of 0 and maximum amplitude of 0 used.

Figure 4. PF-06741086 normalizes hemostasis in moderate FIX deficient whole blood

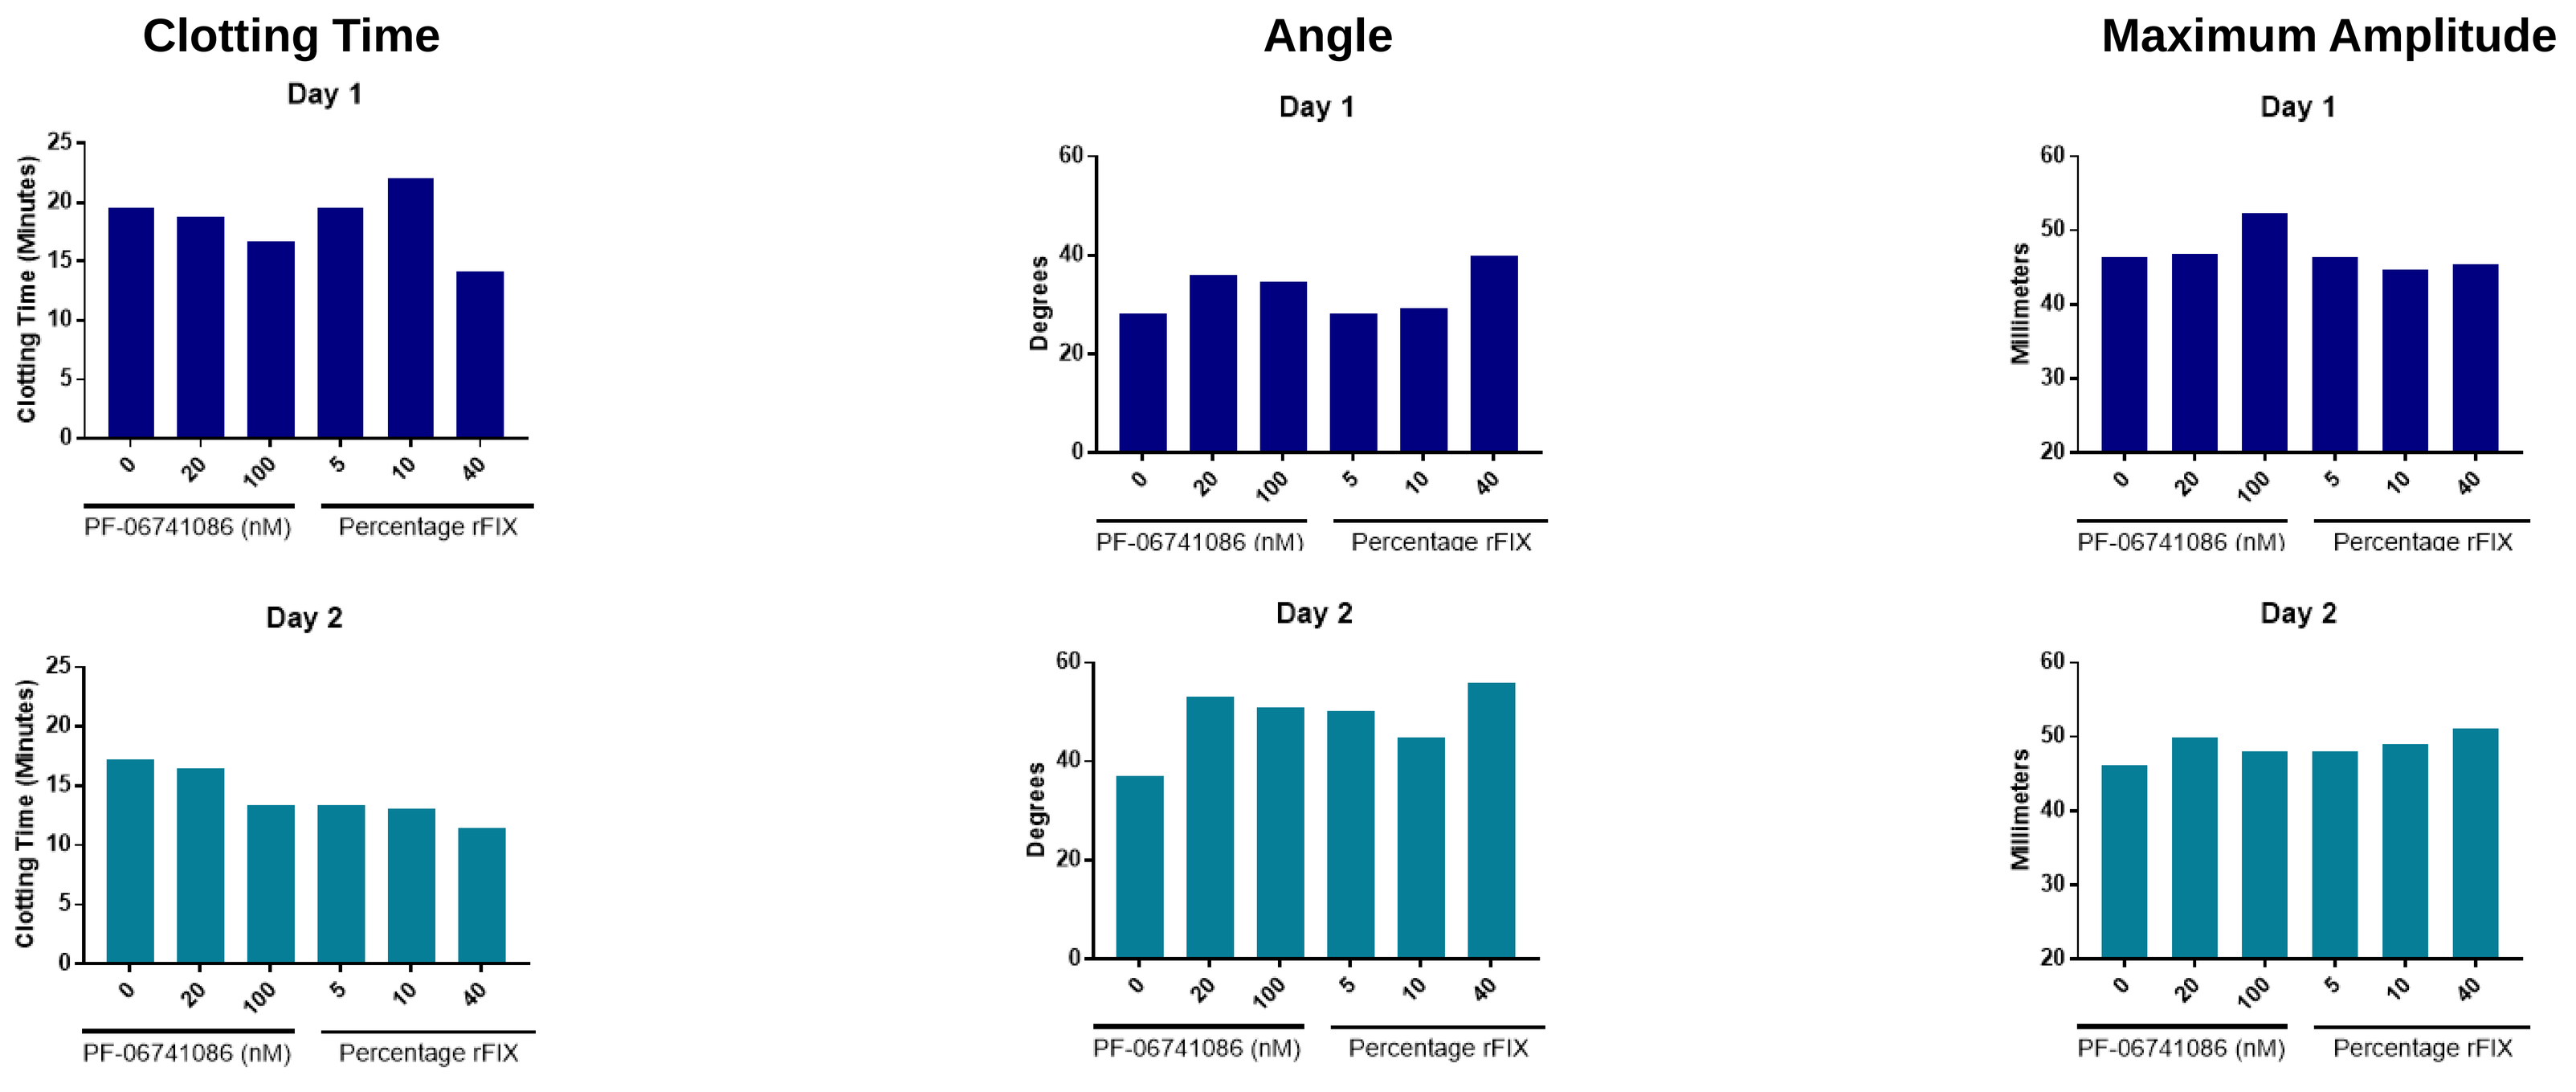
Clotting Time
Angle
Maximum Amplitude
Day 1
Day 2
Clotting Time (Minutes)
Degrees
Millimeters
PF-06741086 (nM)
Percentage rFIX

Figure 4. Response of PF-06741086 in moderate Factor IX Deficient whole blood and plasmas
Ex vivo dosing of PF-06741086 to a final concentration of 20 nM or 100 nM in moderate Factor IX deficient whole blood shows a dose-dependent decrease in clotting time with increases in both angle and amplitude when analyzed by TEG (Day 1) or ROTEM (Day 2). PF-06741086 treatment is observed to induce a pro-coagulant effect comparable to treatment with rFIX.

Figure 5. Log fold changes in hemostatic parameters with addition of PF-06741086 or rFVIII to hemophilia A whole blood

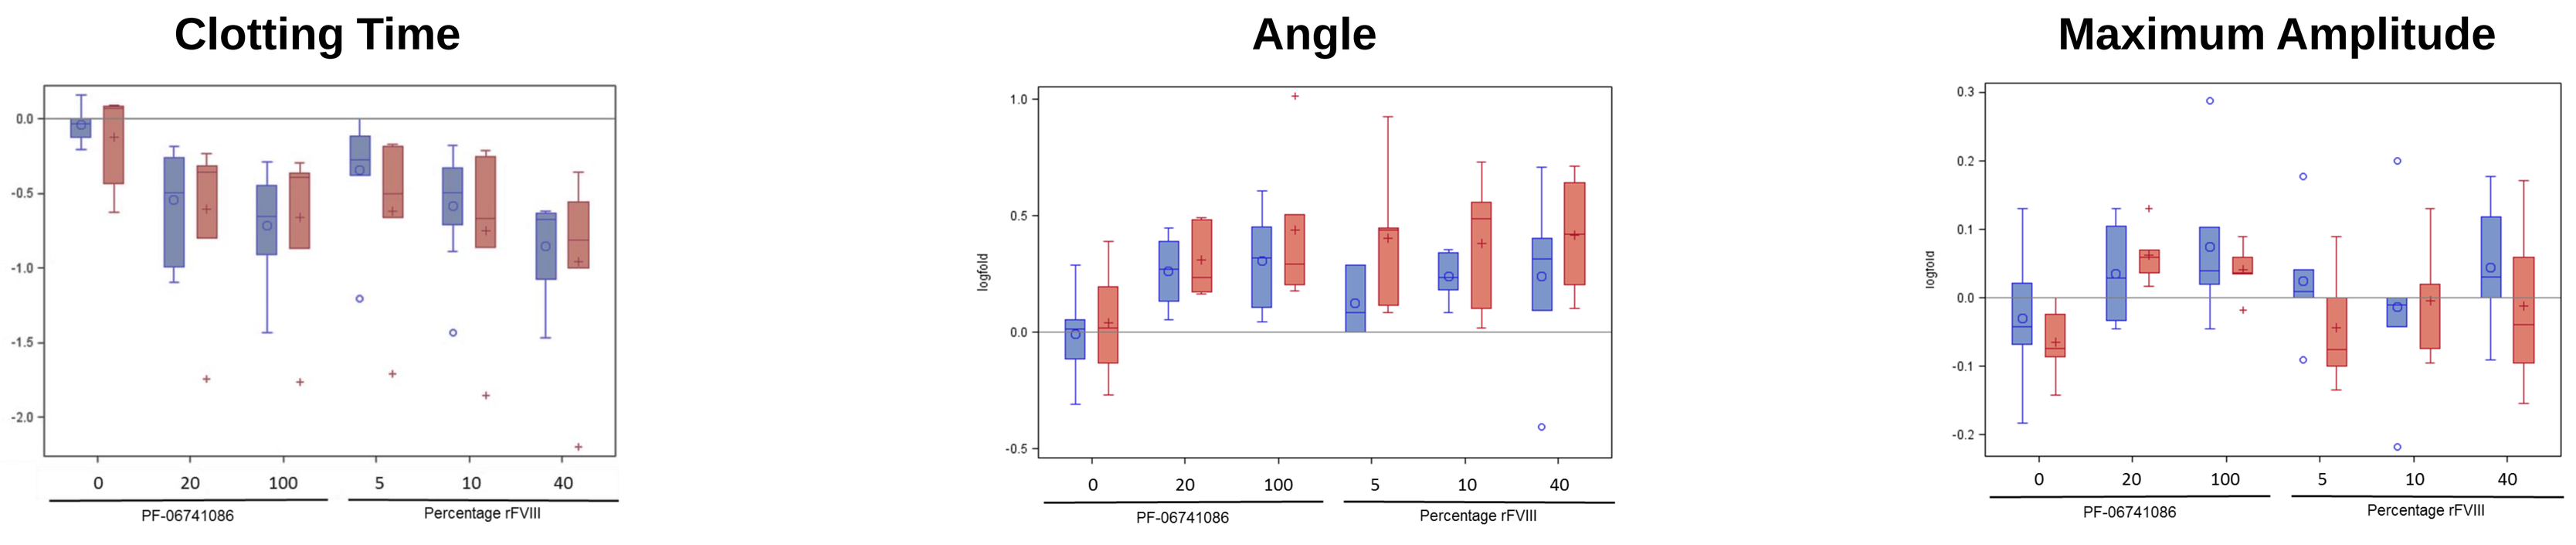
Clotting Time
Angle
Maximum Amplitude
logfold
PF-06741086
Percentage rFVIII

Figure 5. Log-fold changes in hemostatic parameters in response to PF-06741086 or rFVIII in Hemophilia A whole blood
Plots of the log-fold changes in hemostatic parameters following ex vivo treatment of whole blood with PF-06741086 or rFVIII with respect to untreated whole blood from individual Hemophilia A volunteers (n=6) on two separate days (Blue = Day 1 ex vivo dosing n= 9; Red = Day 2 ex vivo dosing, n=7). PF-06741086 shows consistency of response in hemostatic parameters when dosed into whole blood from individual patients on different days.

Conclusions

PF-06741086:

- **Induces reductions in clotting time and increases in angle and amplitude following ex vivo addition to hemophilic whole blood.**
- **Shows consistency of response in whole blood hemostatic assays when dosed into whole blood from the same patient on differing days, despite baseline variability.**
- **Induces pro-coagulant responses in severe FVIII deficient, severe FVIII deficient with FVIII inhibitor and moderate FIX deficient whole blood.**
- **At therapeutically relevant doses, exhibits pro-coagulant activity comparable to addition of recombinant FVIII or FIX in whole blood and plasmas of hemophilia volunteers**
- **Currently in clinical development for treatment of hemophilia A and hemophilia B with and without inhibitors.**

ISTH 2017
1124--PB
Novel Biotherapeutics in Hemophilia
Sunita Patel-Hett
DOI: 10.3252/pso.eu.ISTH2017.2017
Poster presented at ISTH2017 on: Tuesday, July 11
ISTH 2017 CONGRESS BERLIN GERMANY JULY 8-13 WWW.ISTH2017.ORG TRANSCENDING SCIENTIFIC BOUNDARIES
PosterSessionOnline.com